PP09

Automating and Customizing TLG outputs with a Single Click

by Bharathy Prasath and Shweta Padmanaban, Zifo RnD Solutions, Chennai, India

CHALLENGES

1. **Difference in Scope**

 Each sponsor has their own set of requirements for generating TLGs depending on whether it is an input for CSR or for Safety reviews (like DSMB, IB, SMT, DSUR etc.).

2. **The requirements differ in the following aspects**

 Document Type – Individual or Consolidated

 Output Type – rtf, pdf, spreadsheet or all together

 Grouping type – all safety outputs in one document, all tables in one document etc.

3. **Repeated Execution**

 For the above requirements to be fulfilled, programs will have to be executed repeatedly for each type of output.

 Even if the requirements are not complex, running each program individually is still a hectic task.

4. **Titles and Footnotes**

 When the outputs are programmed initially, one of the common things a programmer looks for during self-review is consistency among the titles and footnotes across outputs. Opening each one of the programs or outputs is time consuming.

 A change in titles and footnotes during the interpretation phase is unavoidable. Opening each program to make small changes is time consuming.

5. **Re-runs**

 Depending on the Study, the outputs have to be generated multiple times. It maybe for different data cuts, different stages of safety reviews etc.

 Checking for inappropriate page breaks and creating table of contents for each run takes as much time as developing a table!

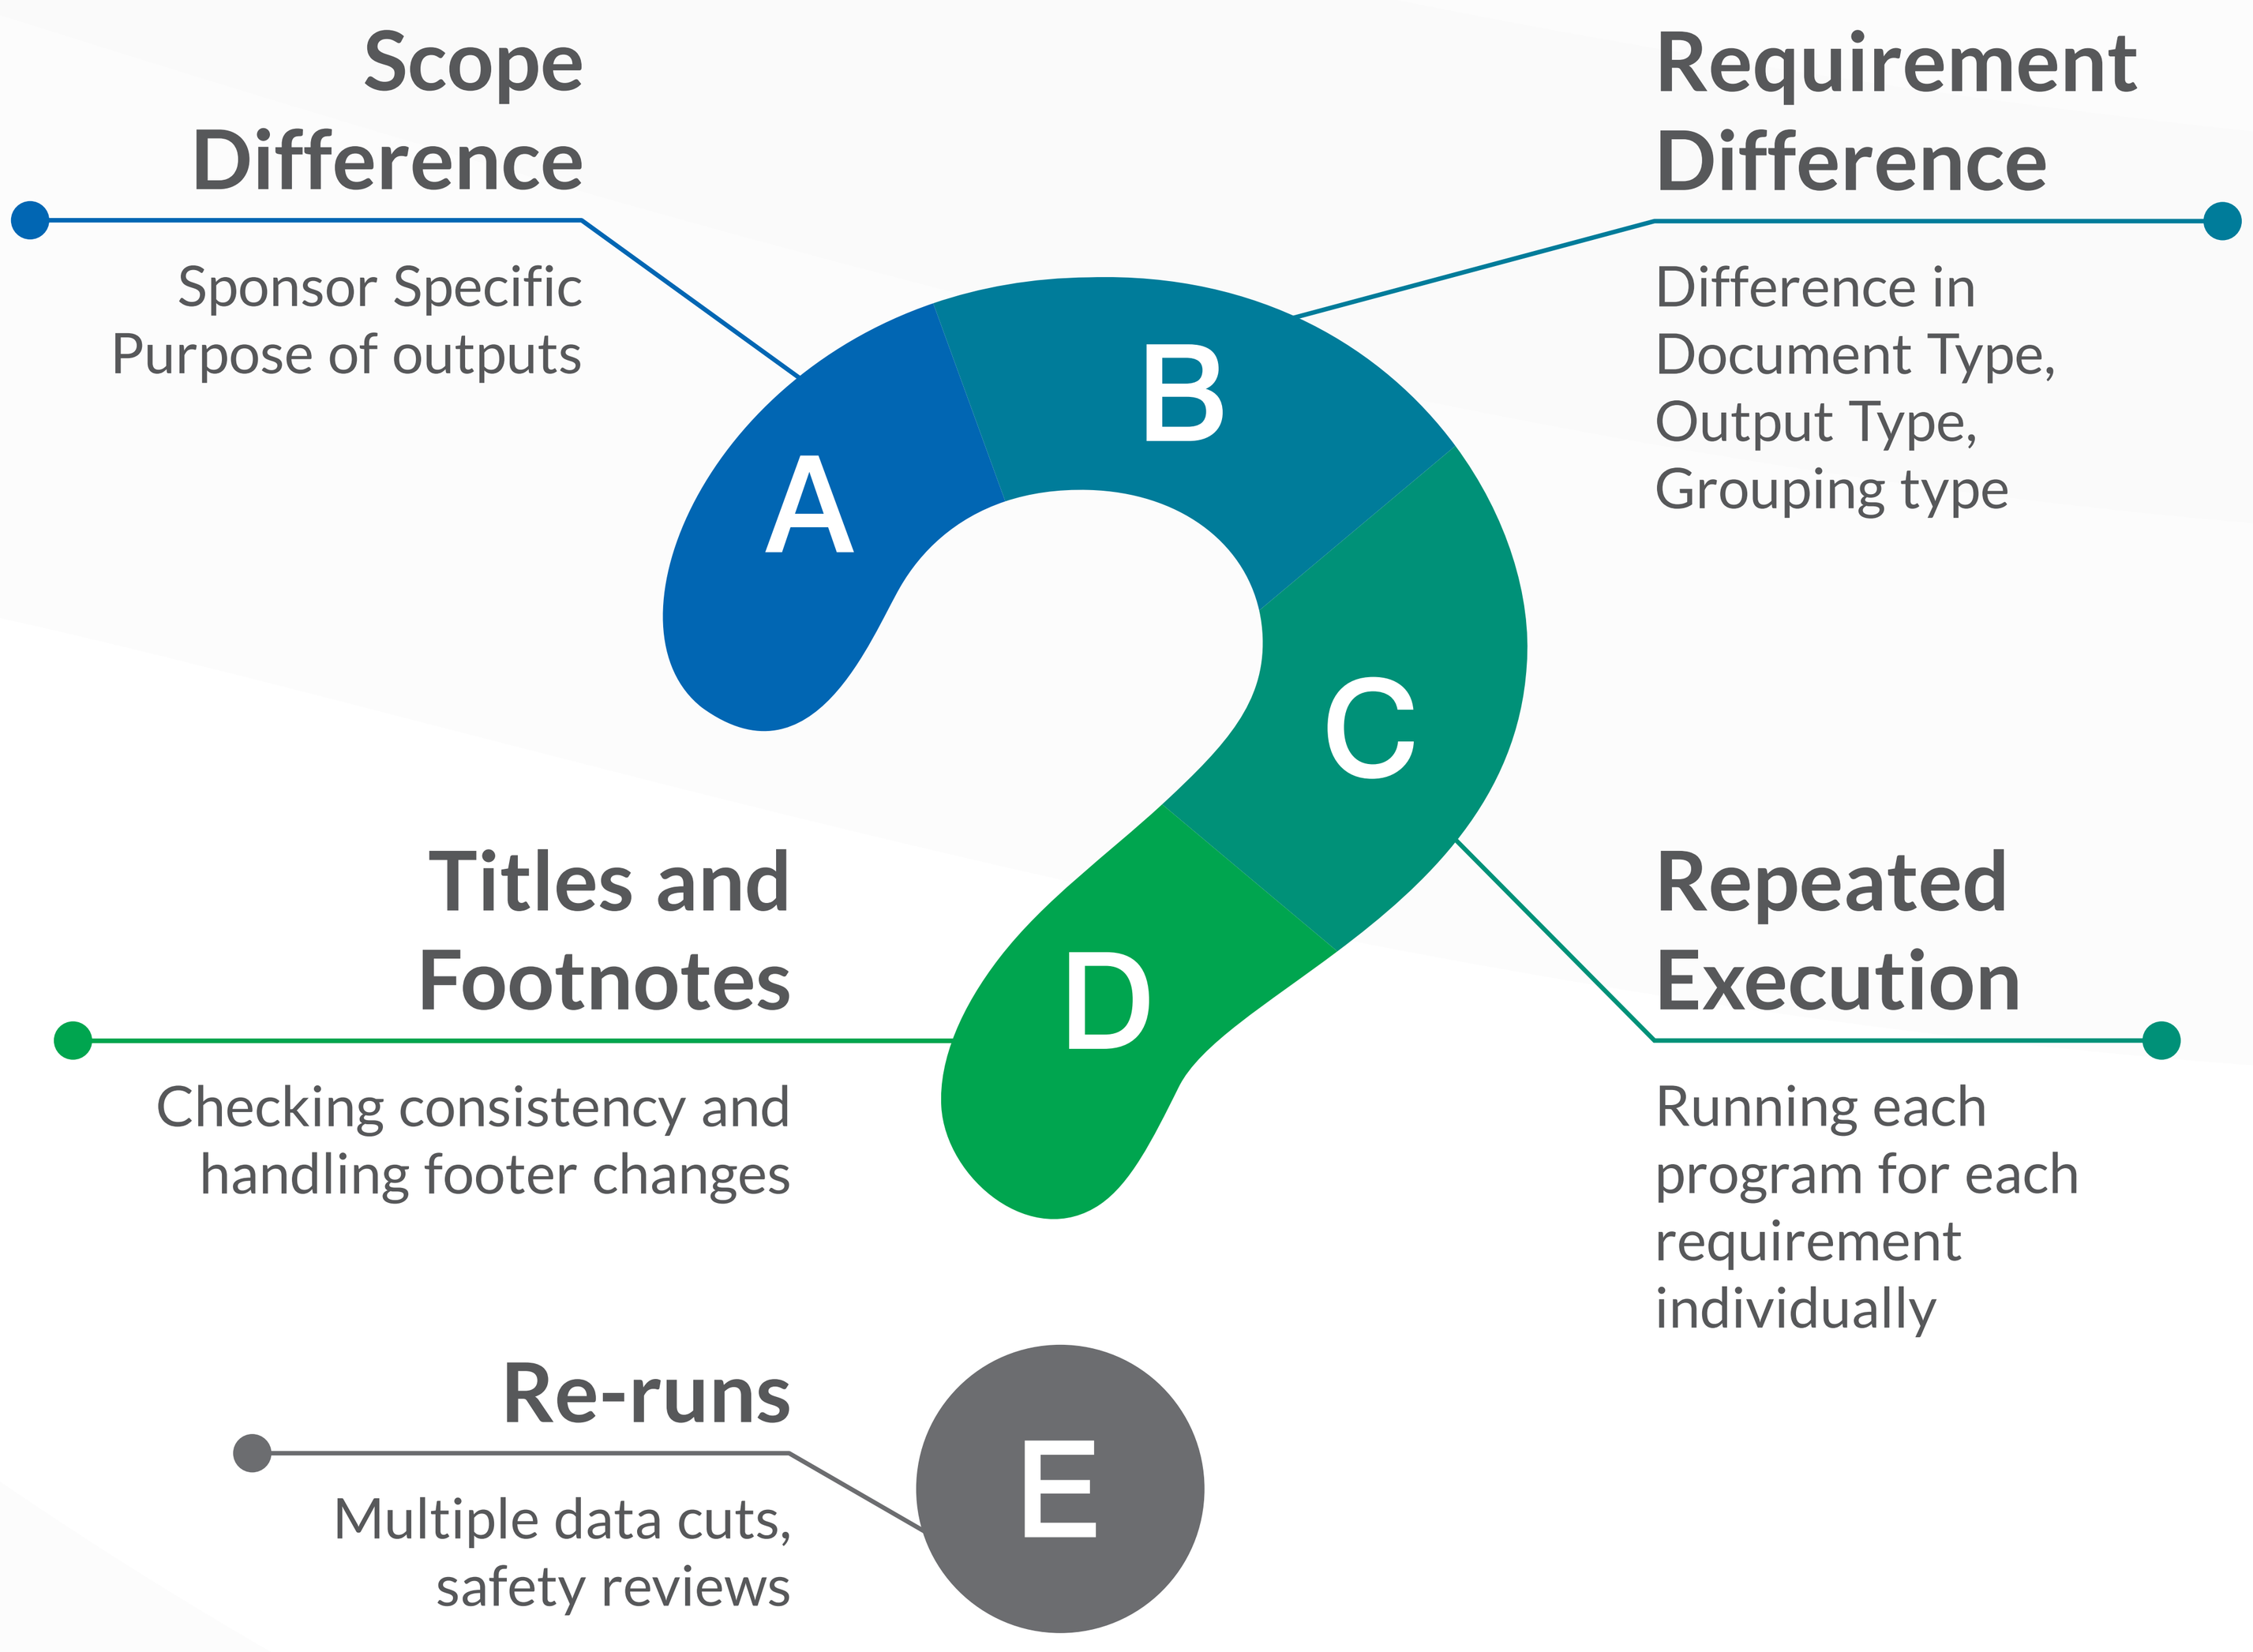
Scope Difference
Sponsor Specific Purpose of outputs
Requirement Difference
Difference in Document Type, Output Type, Grouping type
A
B
C
D
E
Titles and Footnotes
Checking consistency and handling footer changes
Repeated Execution
Running each program for each requirement individually
Re-runs
Multiple data cuts, safety reviews

Contact the Authors at

Bharathy Prasath
bharathy.p@zifornd.com

Shweta Padmanaban
shweta.p@zifornd.com

ZIFO RnD SOLUTIONS

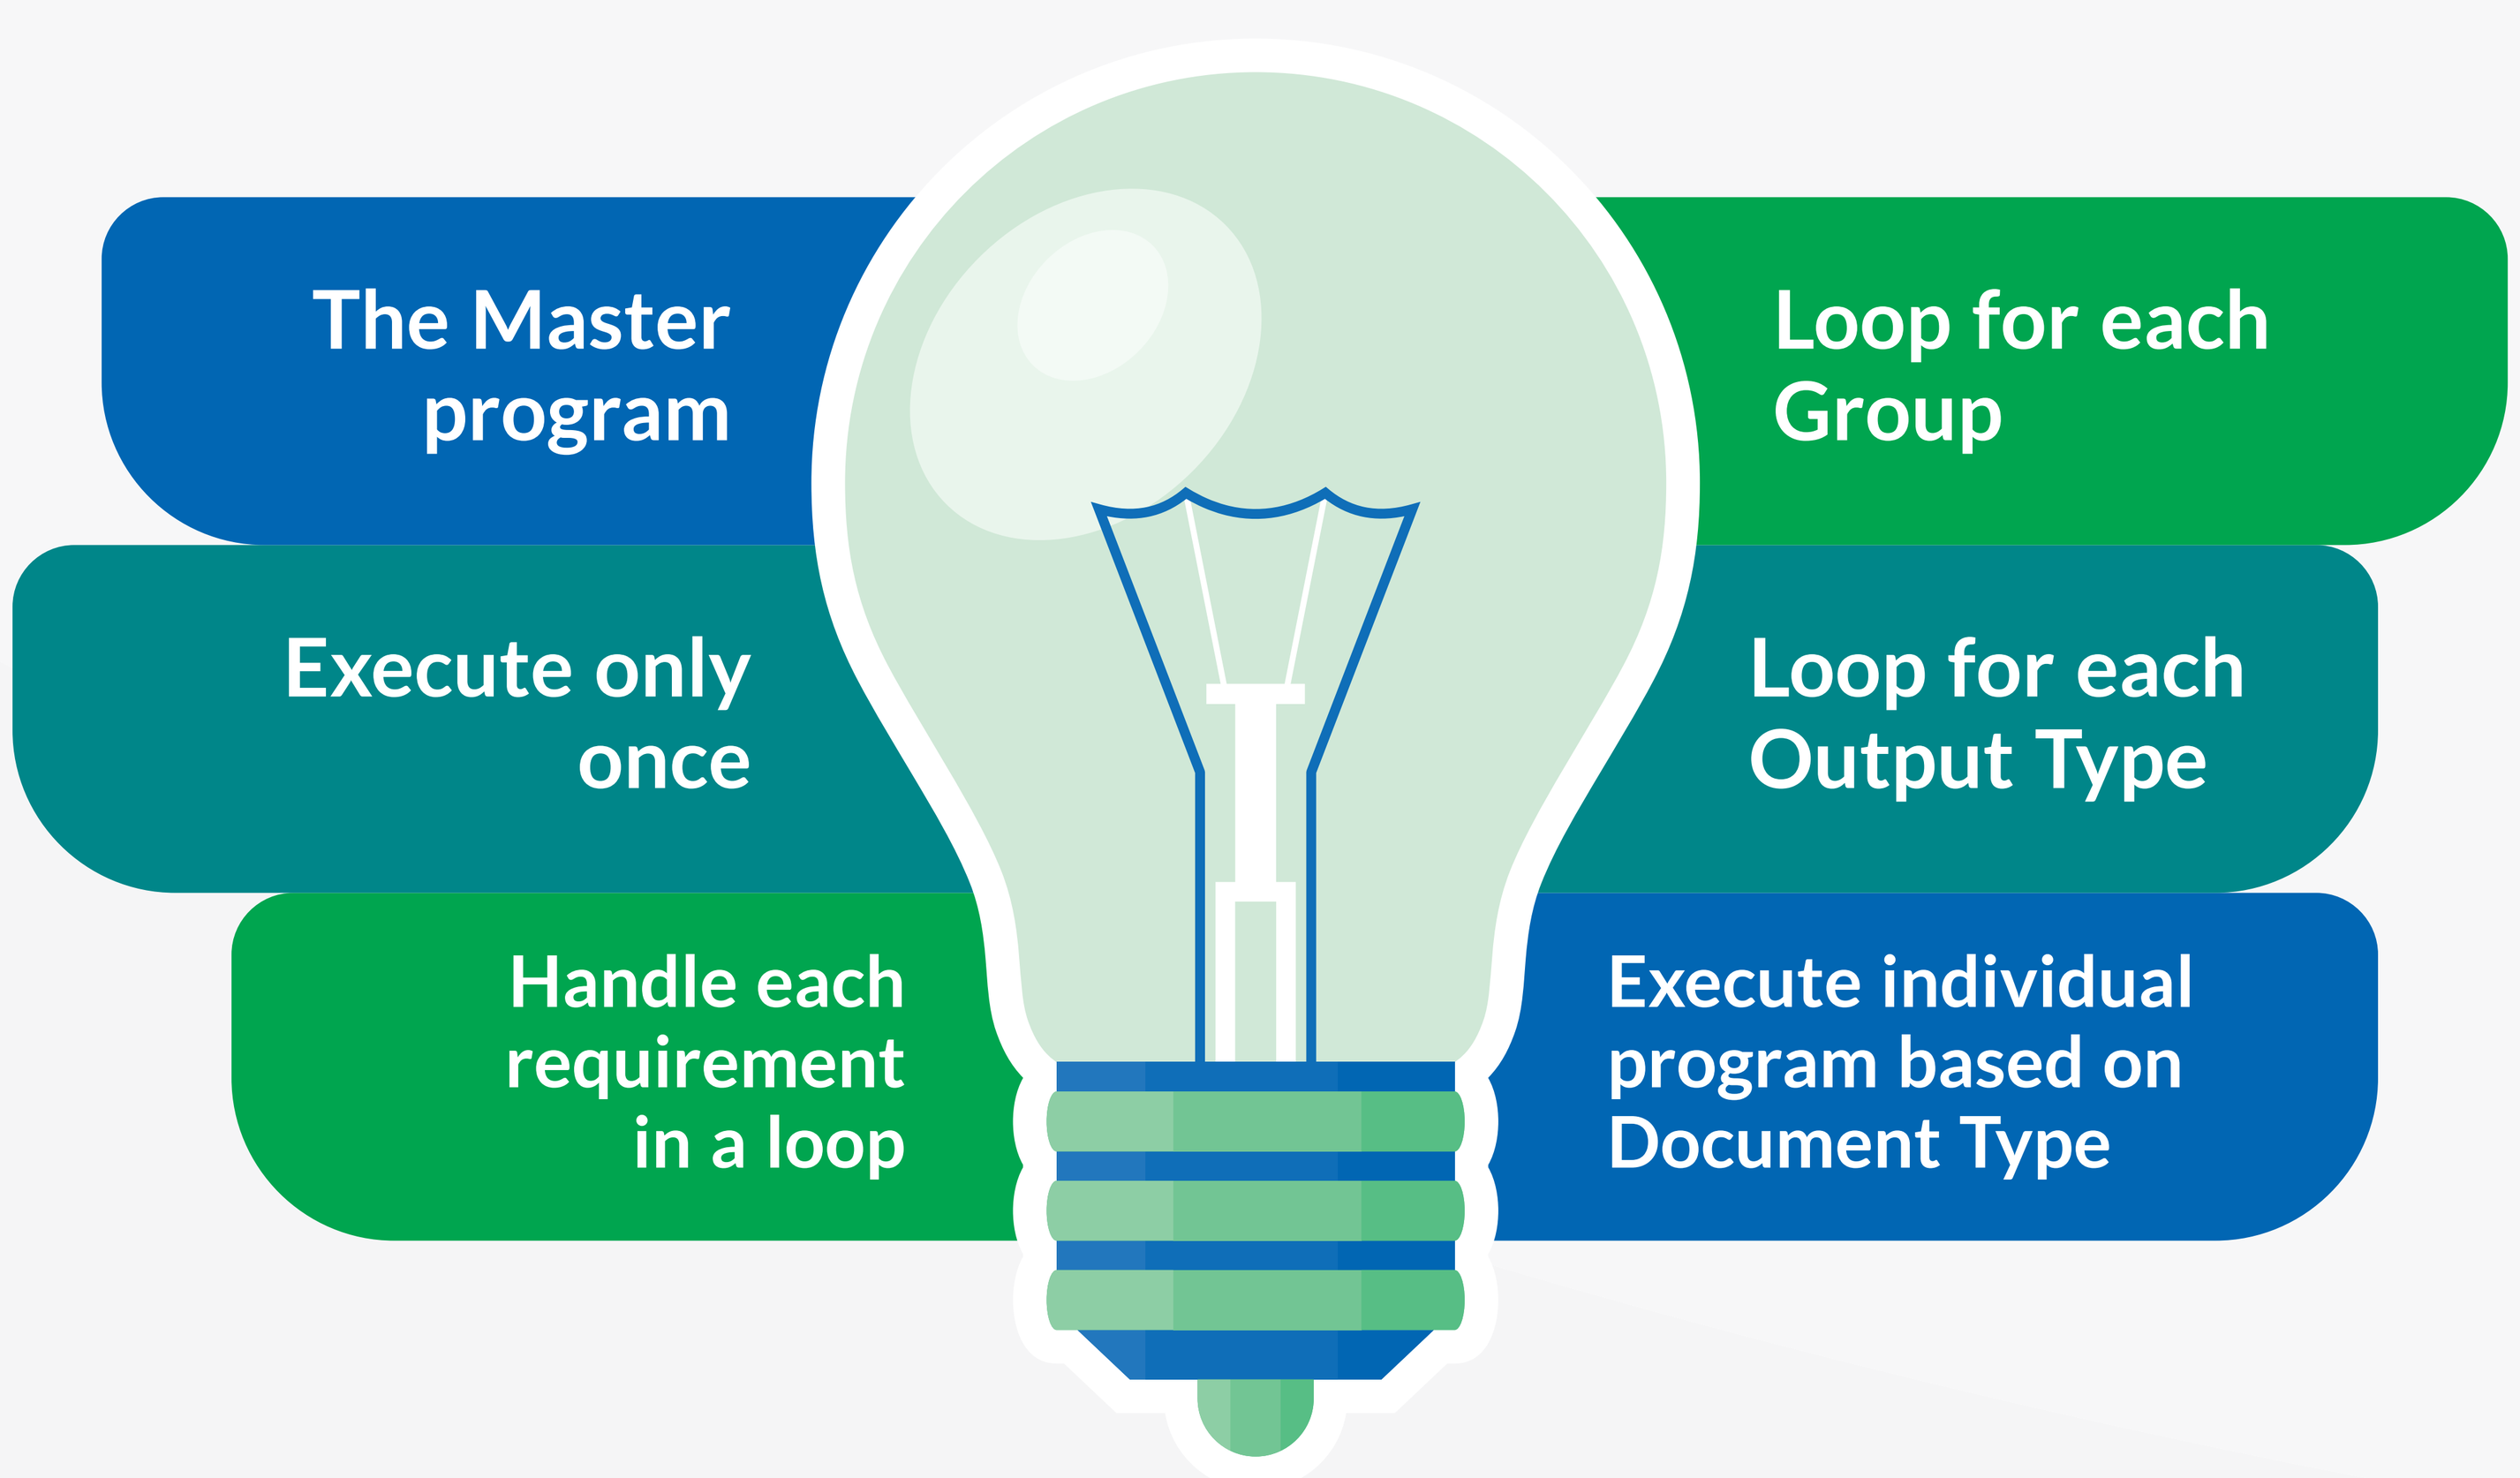
The Master program
Execute only once
Handle each requirement in a loop
Loop for each Group
Loop for each Output Type
Execute individual program based on Document Type

ACCELERATORS

1. **The titles and footnotes handler**

 All the titles and footnotes can be added into one master document.

 This document can be imported and used in programs.

 Whenever there is a change in titles or footnotes, only the master document needs to be updated and the programs remain untouched. This way, only the master document needs re-validation.

2. **Remove page breaks**

 In certain cases, while printing data on an rtf file, additional page breaks might be generated. The % removesection SAS macro reads the rtf document and removes additional page breaks.

3. **Error Warning Log**

 Executing each program and checking for log issues manually is time consuming and to be honest, a little boring to scroll through.

 The % errwarnlog SAS macro gives out error, warning and forbidden note counts based on the outputs and issue categories. It also prints fragments of the SAS log which have issues.

SOLUTION

1. **The Master program**

 Individual programs can be included into one program so that everything can be executed in one go.

2. **Handling different Requirements**

 a. *Depending on the required number of Groups and number of outputs in each group, individual programs can be combined and executed in the master program.*

 b. *Within each Group, depending on the document type (individual or consolidated), programs can be executed in loops.*

 c. *The whole process has to be repeated for each output type (rtf, pdf or both). This is not done along with the above methods since each output type requires different SAS features and options.*

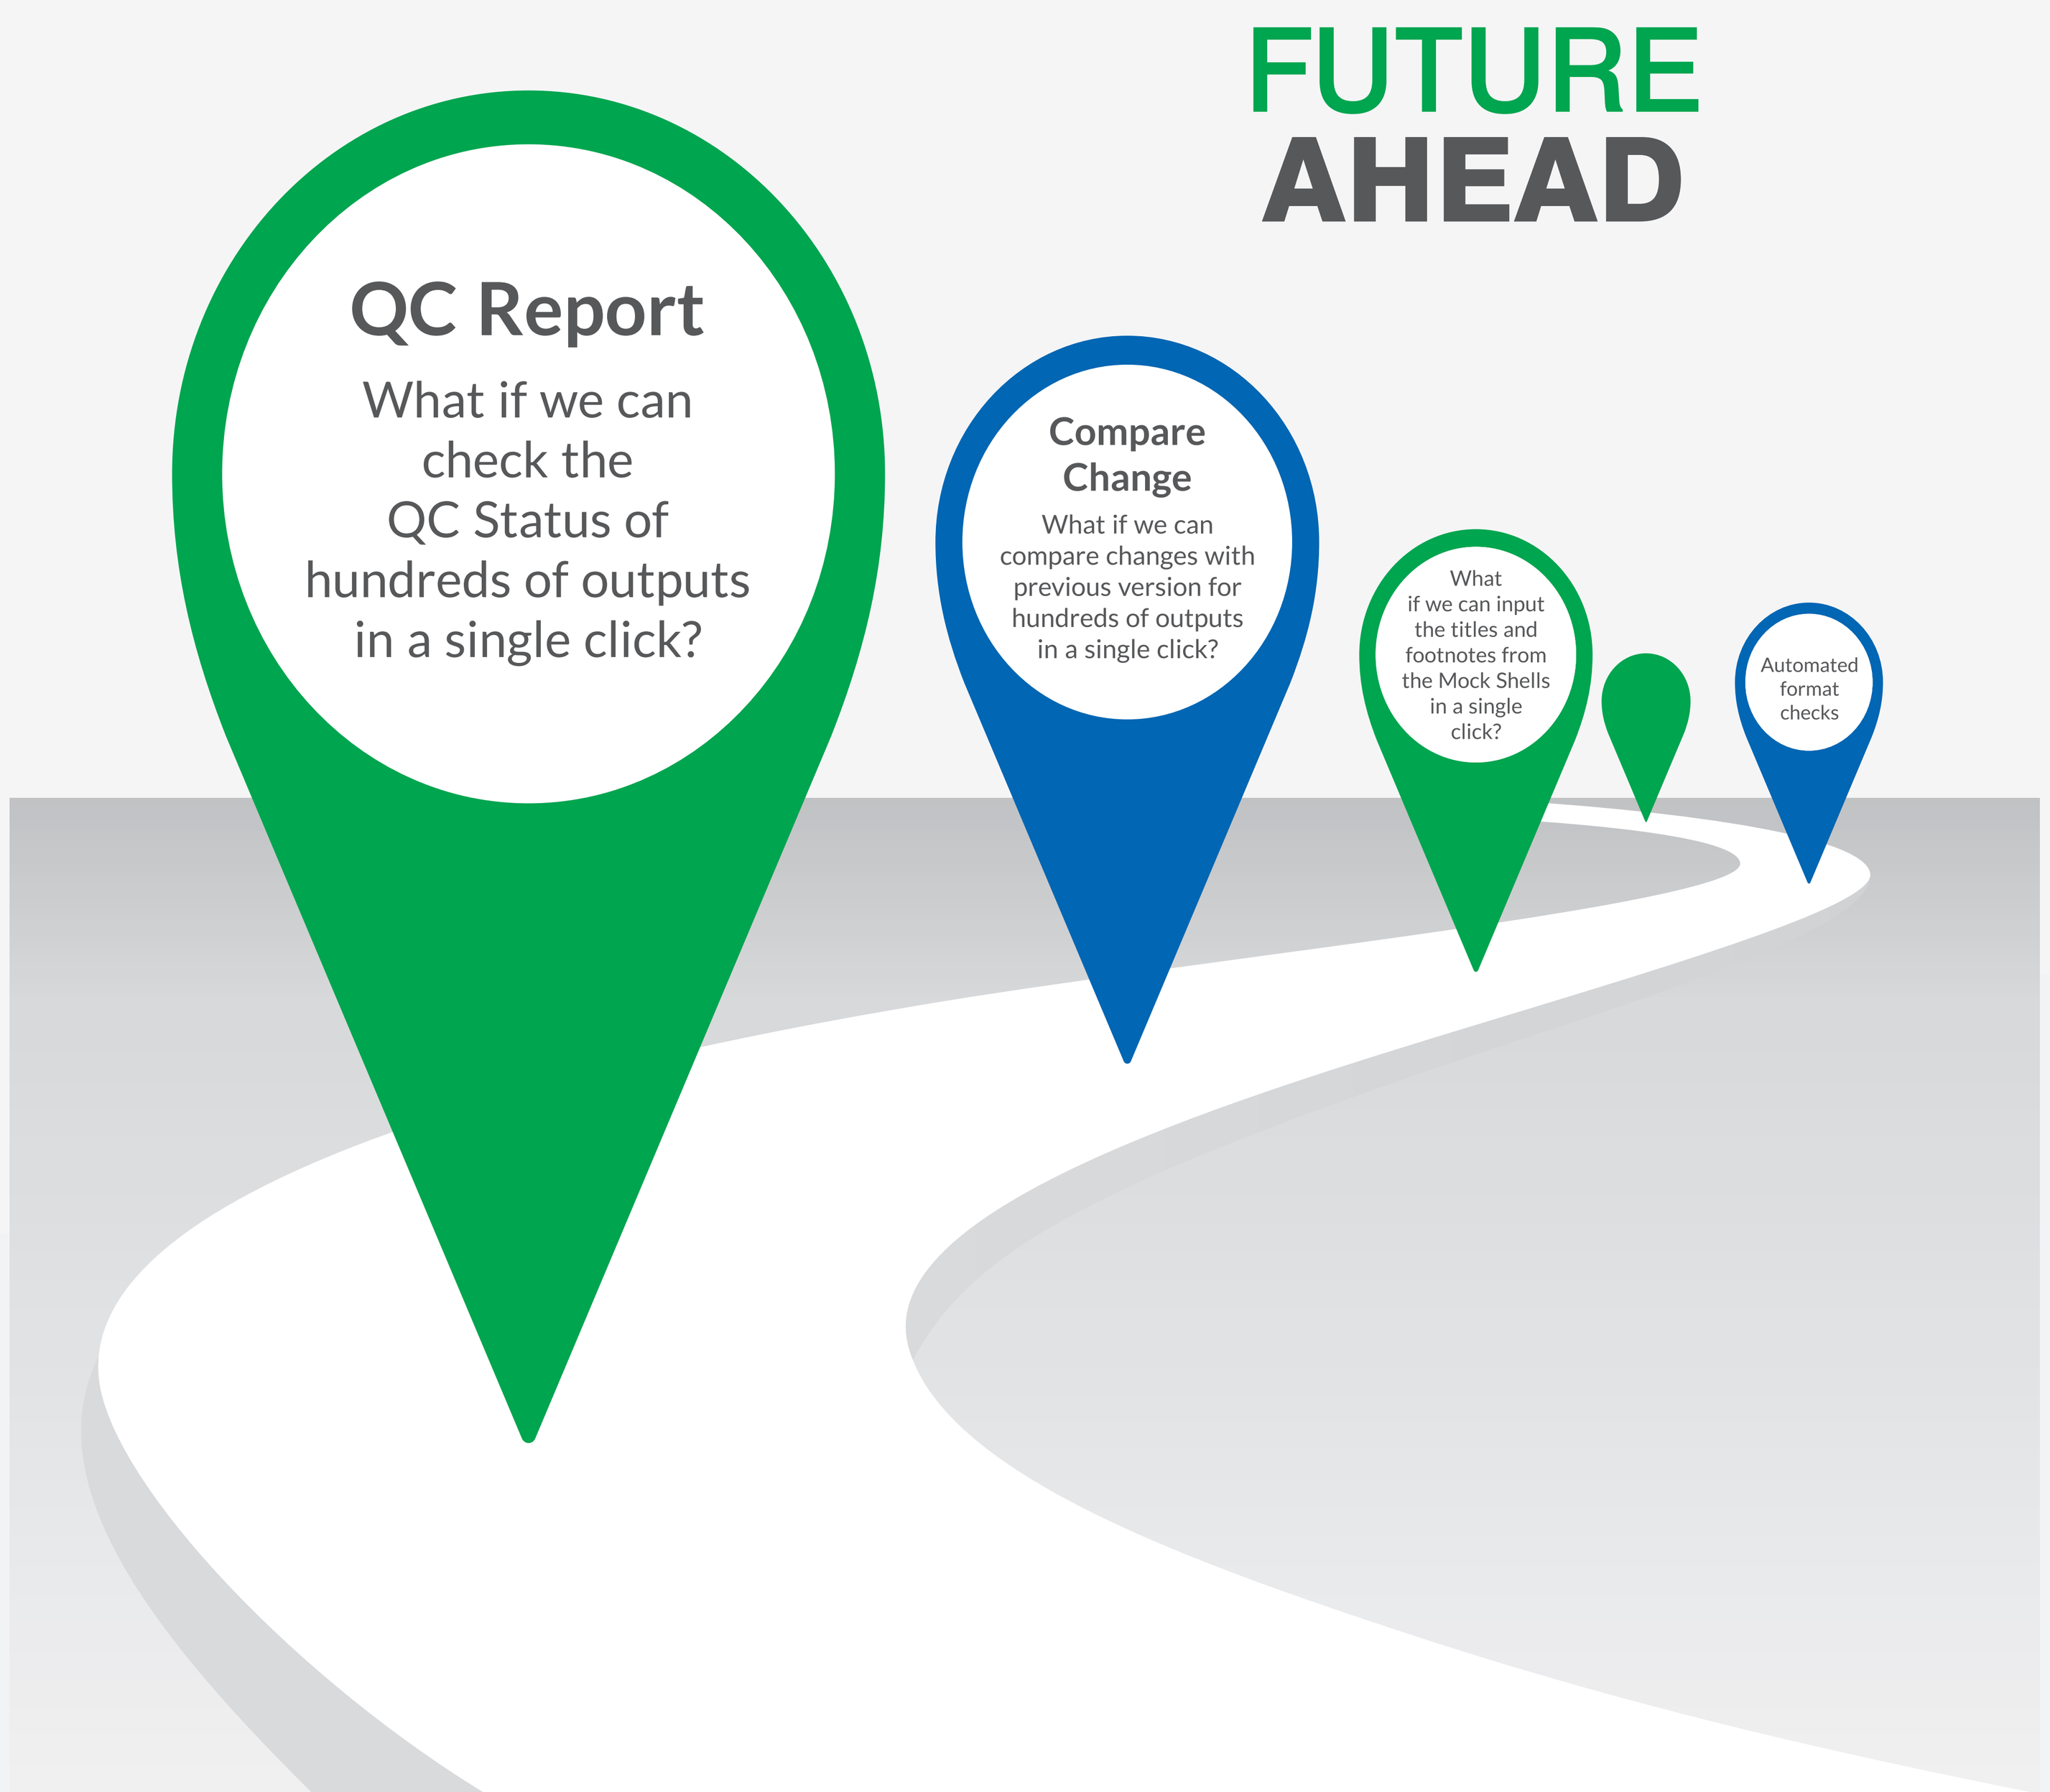
FUTURE
AHEAD
QC Report
What if we can check the QC Status of hundreds of outputs in a single click?
Compare Change
What if we can compare changes with previous version for hundreds of outputs in a single click?
What if we can input the titles and footnotes from the Mock Shells in a single click?
Automated format checks