THE CLINICAL RESEARCH TRAGEDY

Scott Bahlavooni **Chris Decker** **Chris Connor**

"Increasing the pool of researchers who can access data and decreasing the time it takes for them to review and find new patterns in that data is critical to speeding up development of lifesaving treatments for patients."

Joe Biden, Former Vice President, U.S.

SELF-INFLICTED COMPLEXITY

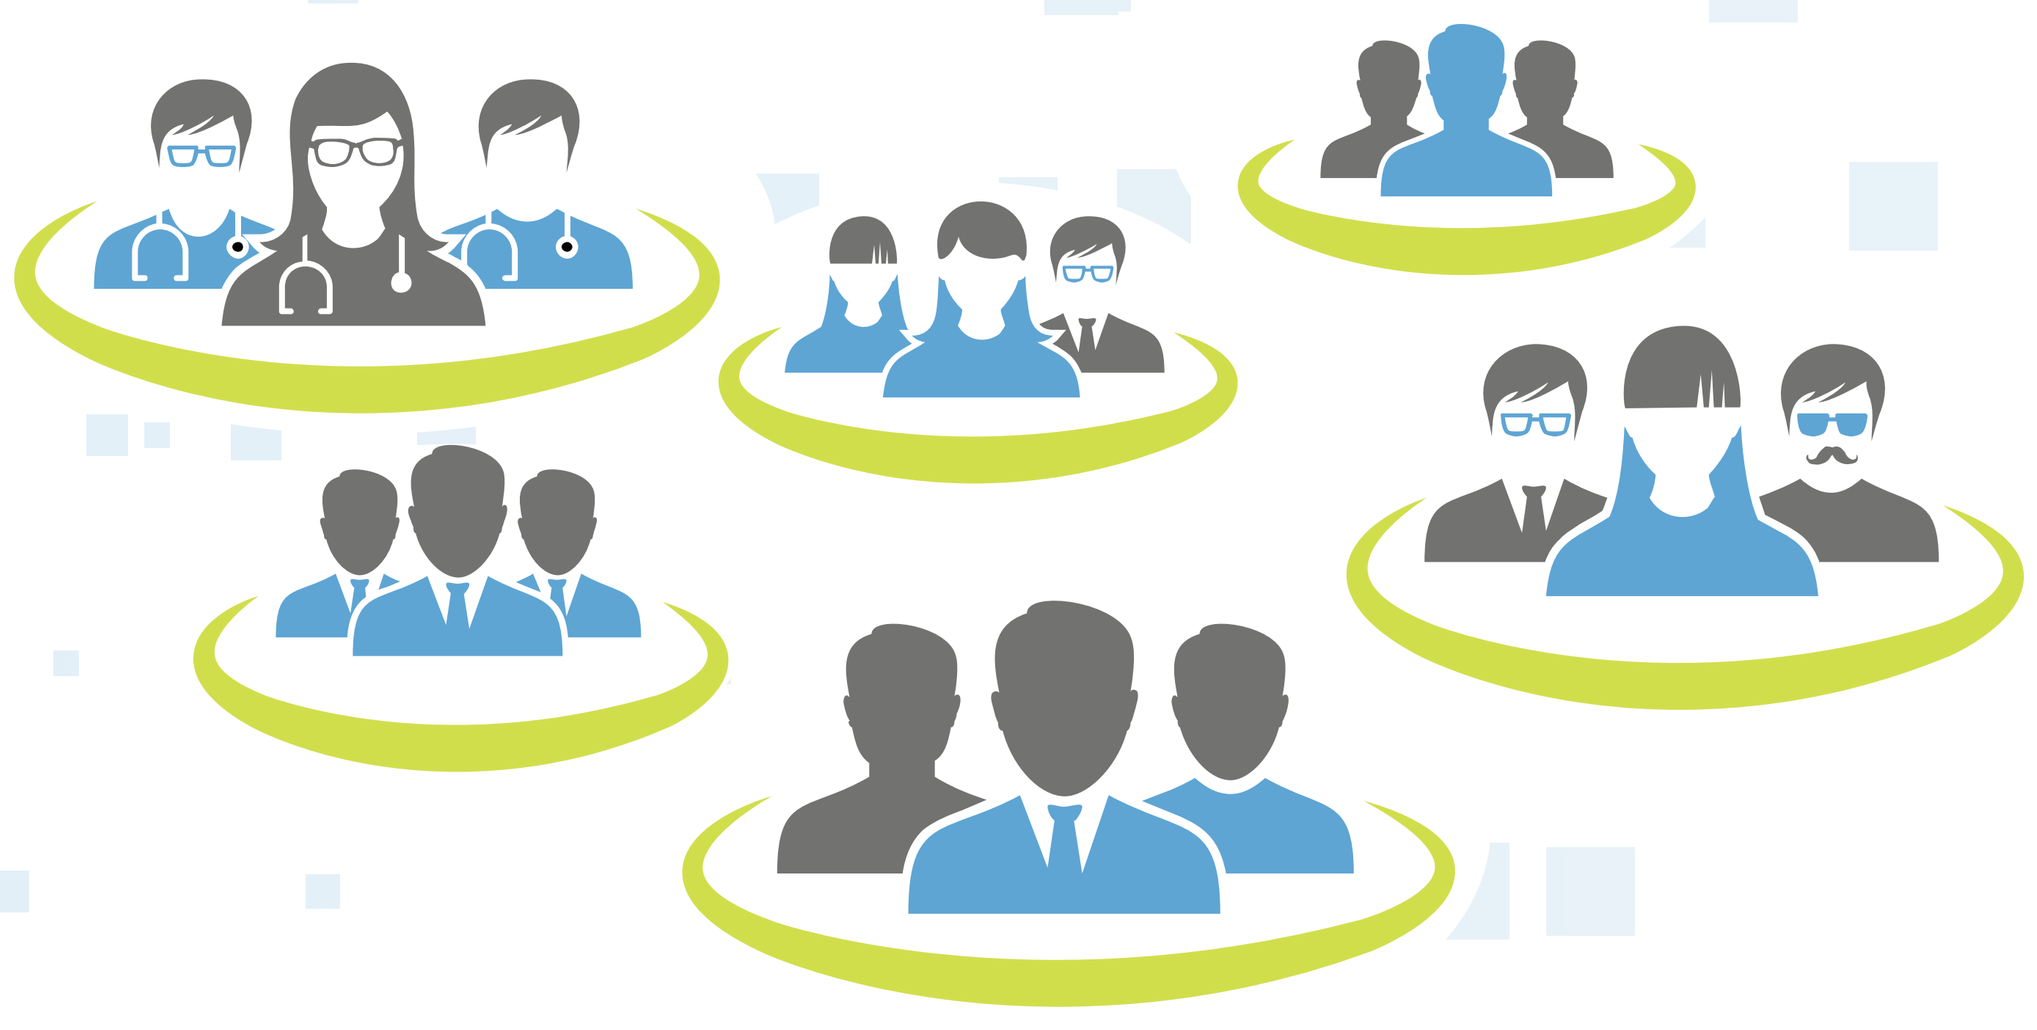

- Siloed neighborhoods lack understanding of customer's needs
- Many manual handoffs increase time and reduce quality
- Tower of Babel: disconnected languages, tools, and processes

REGULATORY HURDLES

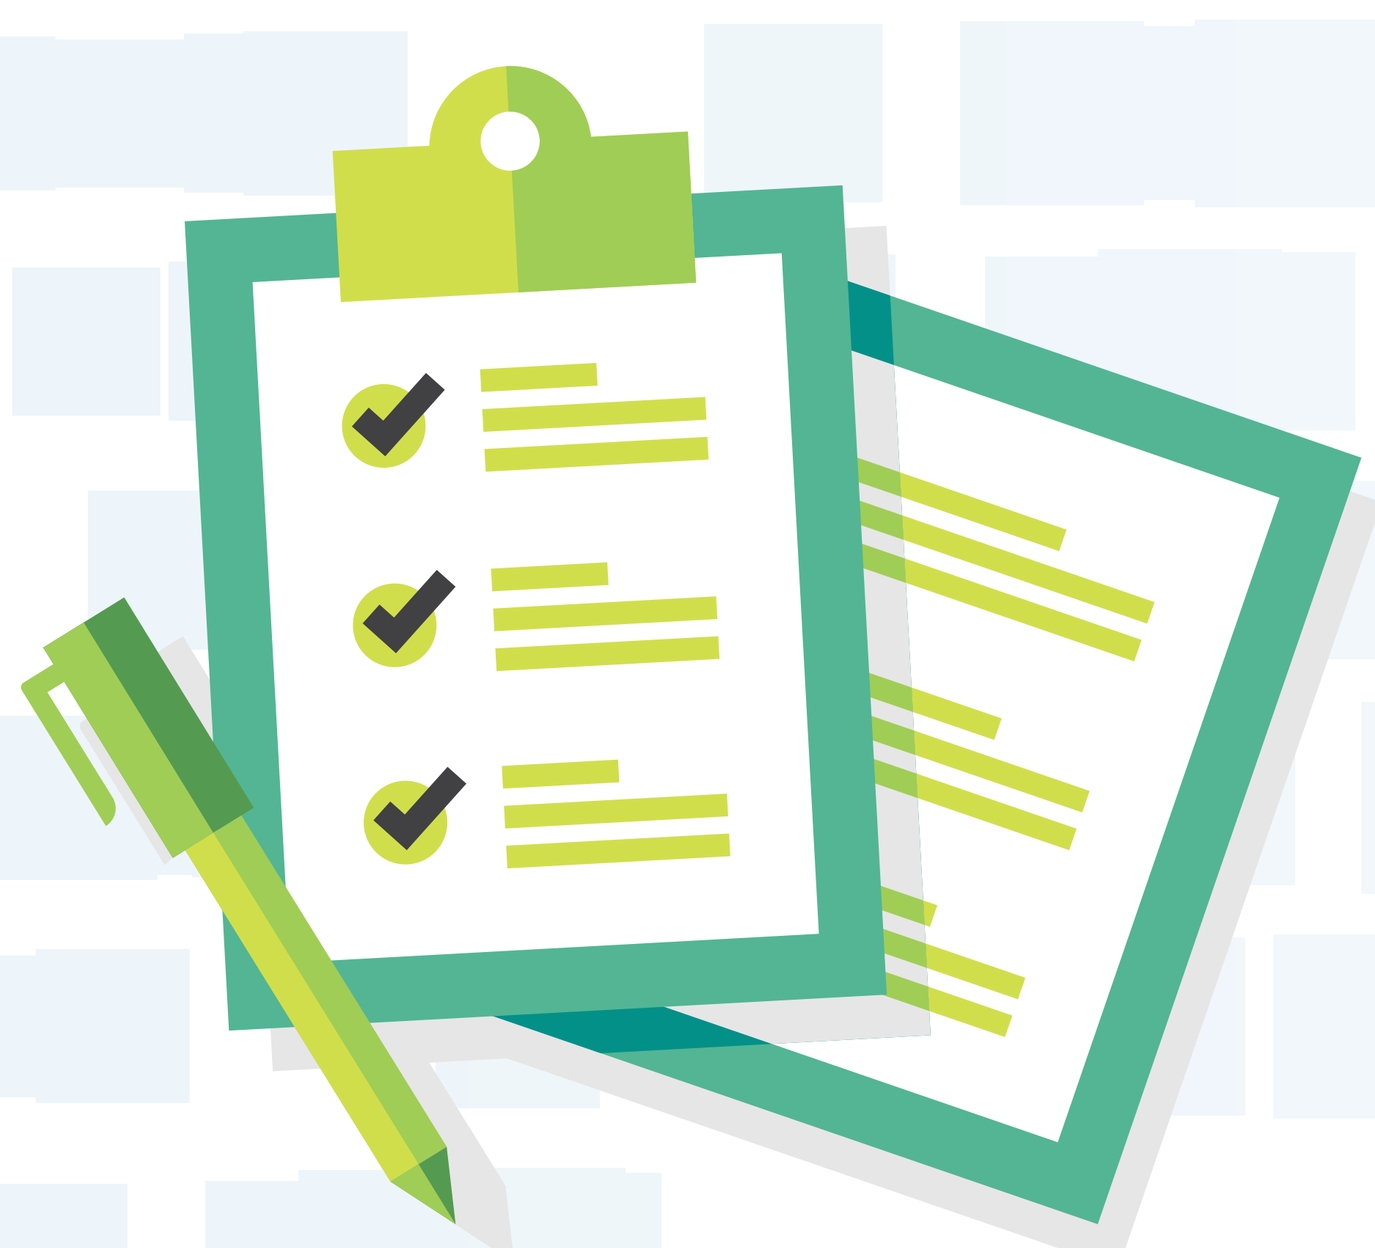

- Submission gauntlet that penalizes innovation at every step
- Piles of black box, vague, and confusing regulations
- Rigidness, risk averse, and fear of change

DATA HERE, THERE, & EVERYWHERE

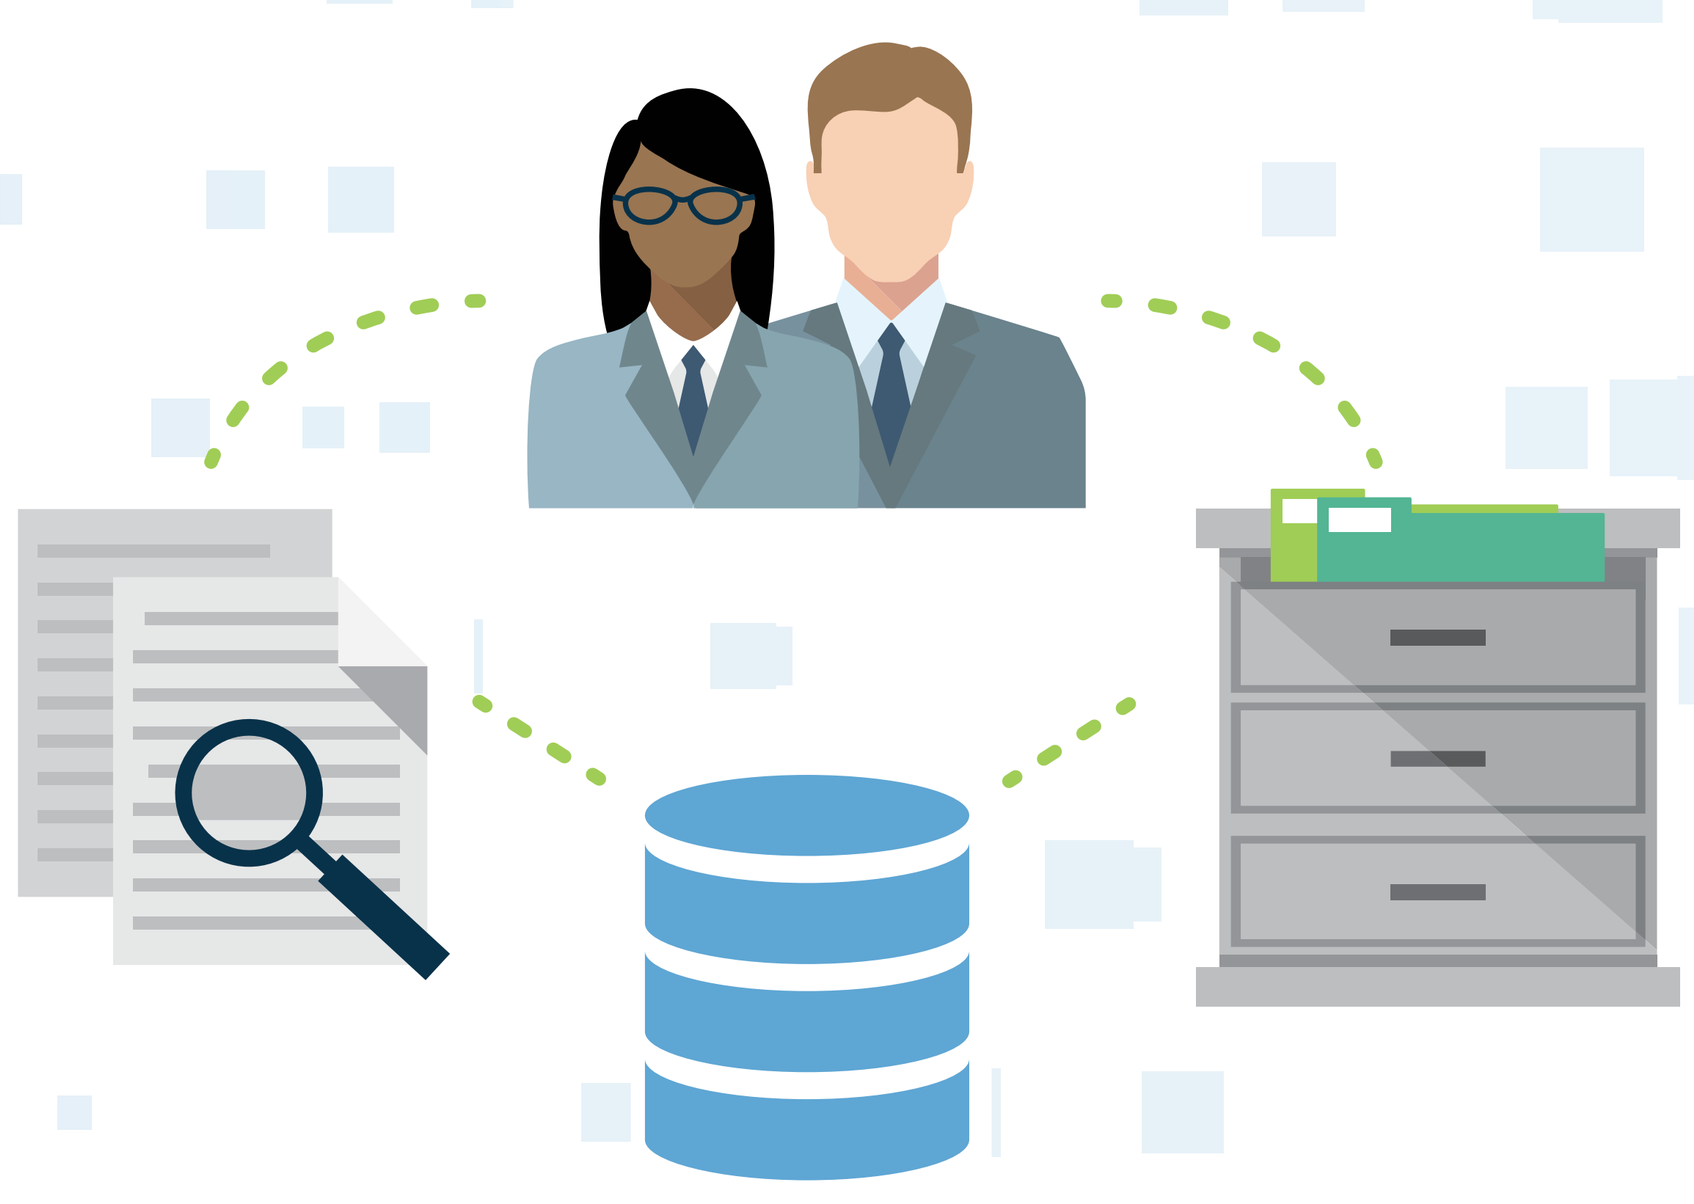

- Collection data, raw data, SDTM data, ADaM data, your data, my data
- Copies and copies and copies of data for every milestone
- Misperception that quality data and traceability exist

DATA DEVOID OF CONTEXT

ag.xpt

Row	STUDYID	DOMAIN	USUBJID	AGSEQ	AGLNKID	AGTRT	AGCAT	AGDOSE	AGDOSEU	AGSTDTC
1	ABC123	AG	AD01-101	1	03	18F-Florbetapir	AMYLOID TRACER	370	MBq	2012-05-22T08:40:00
2	ABC123	AG	AD01-102	1	04	11C-PiB	AMYLOID TRACER	370	MBq	2012-05-22T07:20:00
3	ABC123	AG	AD01-103	1	05	FDG	GLUCOSE TRACER	400	MBq	2012-05-22T08:30:00

pr.xpt

Row	STUDYID	DOMAIN	USUBJID	SPDEVID	PRSEQ	PRLNKID	PRTRT	PRLOC	PRFAST	PRSTDTC
1	ABC123	PR	AD01-101	22	1	03	PET/CT	HEAD	Y	2012-05-22T09:30:00
2	ABC123	PR	AD01-102	22	1	04	PET/CT	HEAD	Y	2012-05-22T08:00:00
3	ABC123	PR	AD01-103	44	1	05	PET	HEAD	Y	2012-05-22T09:00:00

mo.xpt

Row	STUDYID	DOMAIN	USUBJID	SPDEVID	MOSEQ	MOLNKID	MOREFID	MOTESTCD	MOTEST	MOORRES	MOORRESU	MOSTRESC	MOSTRESN	MOSTRESU
1	ABC123	MO	AD01-101	16	1	02	1234	VOLUME	Volume	1.8	mL	1.8	1.8	mL
2	ABC123	MO	AD01-101	16	2	02	1234	VOLUME	Volume	1.9	mL	1.9	1.9	mL
3	ABC123	MO	AD01-101	16	3	02	1234	VOLUME	Volume	3.7	mL	3.7	3.7	mL
4	ABC123	MO	AD01-101	16	4	02	1234	THICK	Thickness	3	mm	3	3	mm
5	ABC123	MO	AD01-101	16	5	02	1235	VOLUME	Volume	864	mL	864	864	mL

- Clinical information is complex, but our data is two dimensional
- Relationships are in people's heads and not in their data
- Misdirected focus on the transformation and not on the patient

SOLVING 21ST CENTURY PROBLEMS WITH 20TH CENTURY TOOLS

"In the real world, we want to capture the complexity and diversity of the standard of care without forcing it into an artificial structure. The current framework forces researchers to constrain data to a structure that is not intuitive and limits the value of the data."

Florence Barkats, RWE Expert

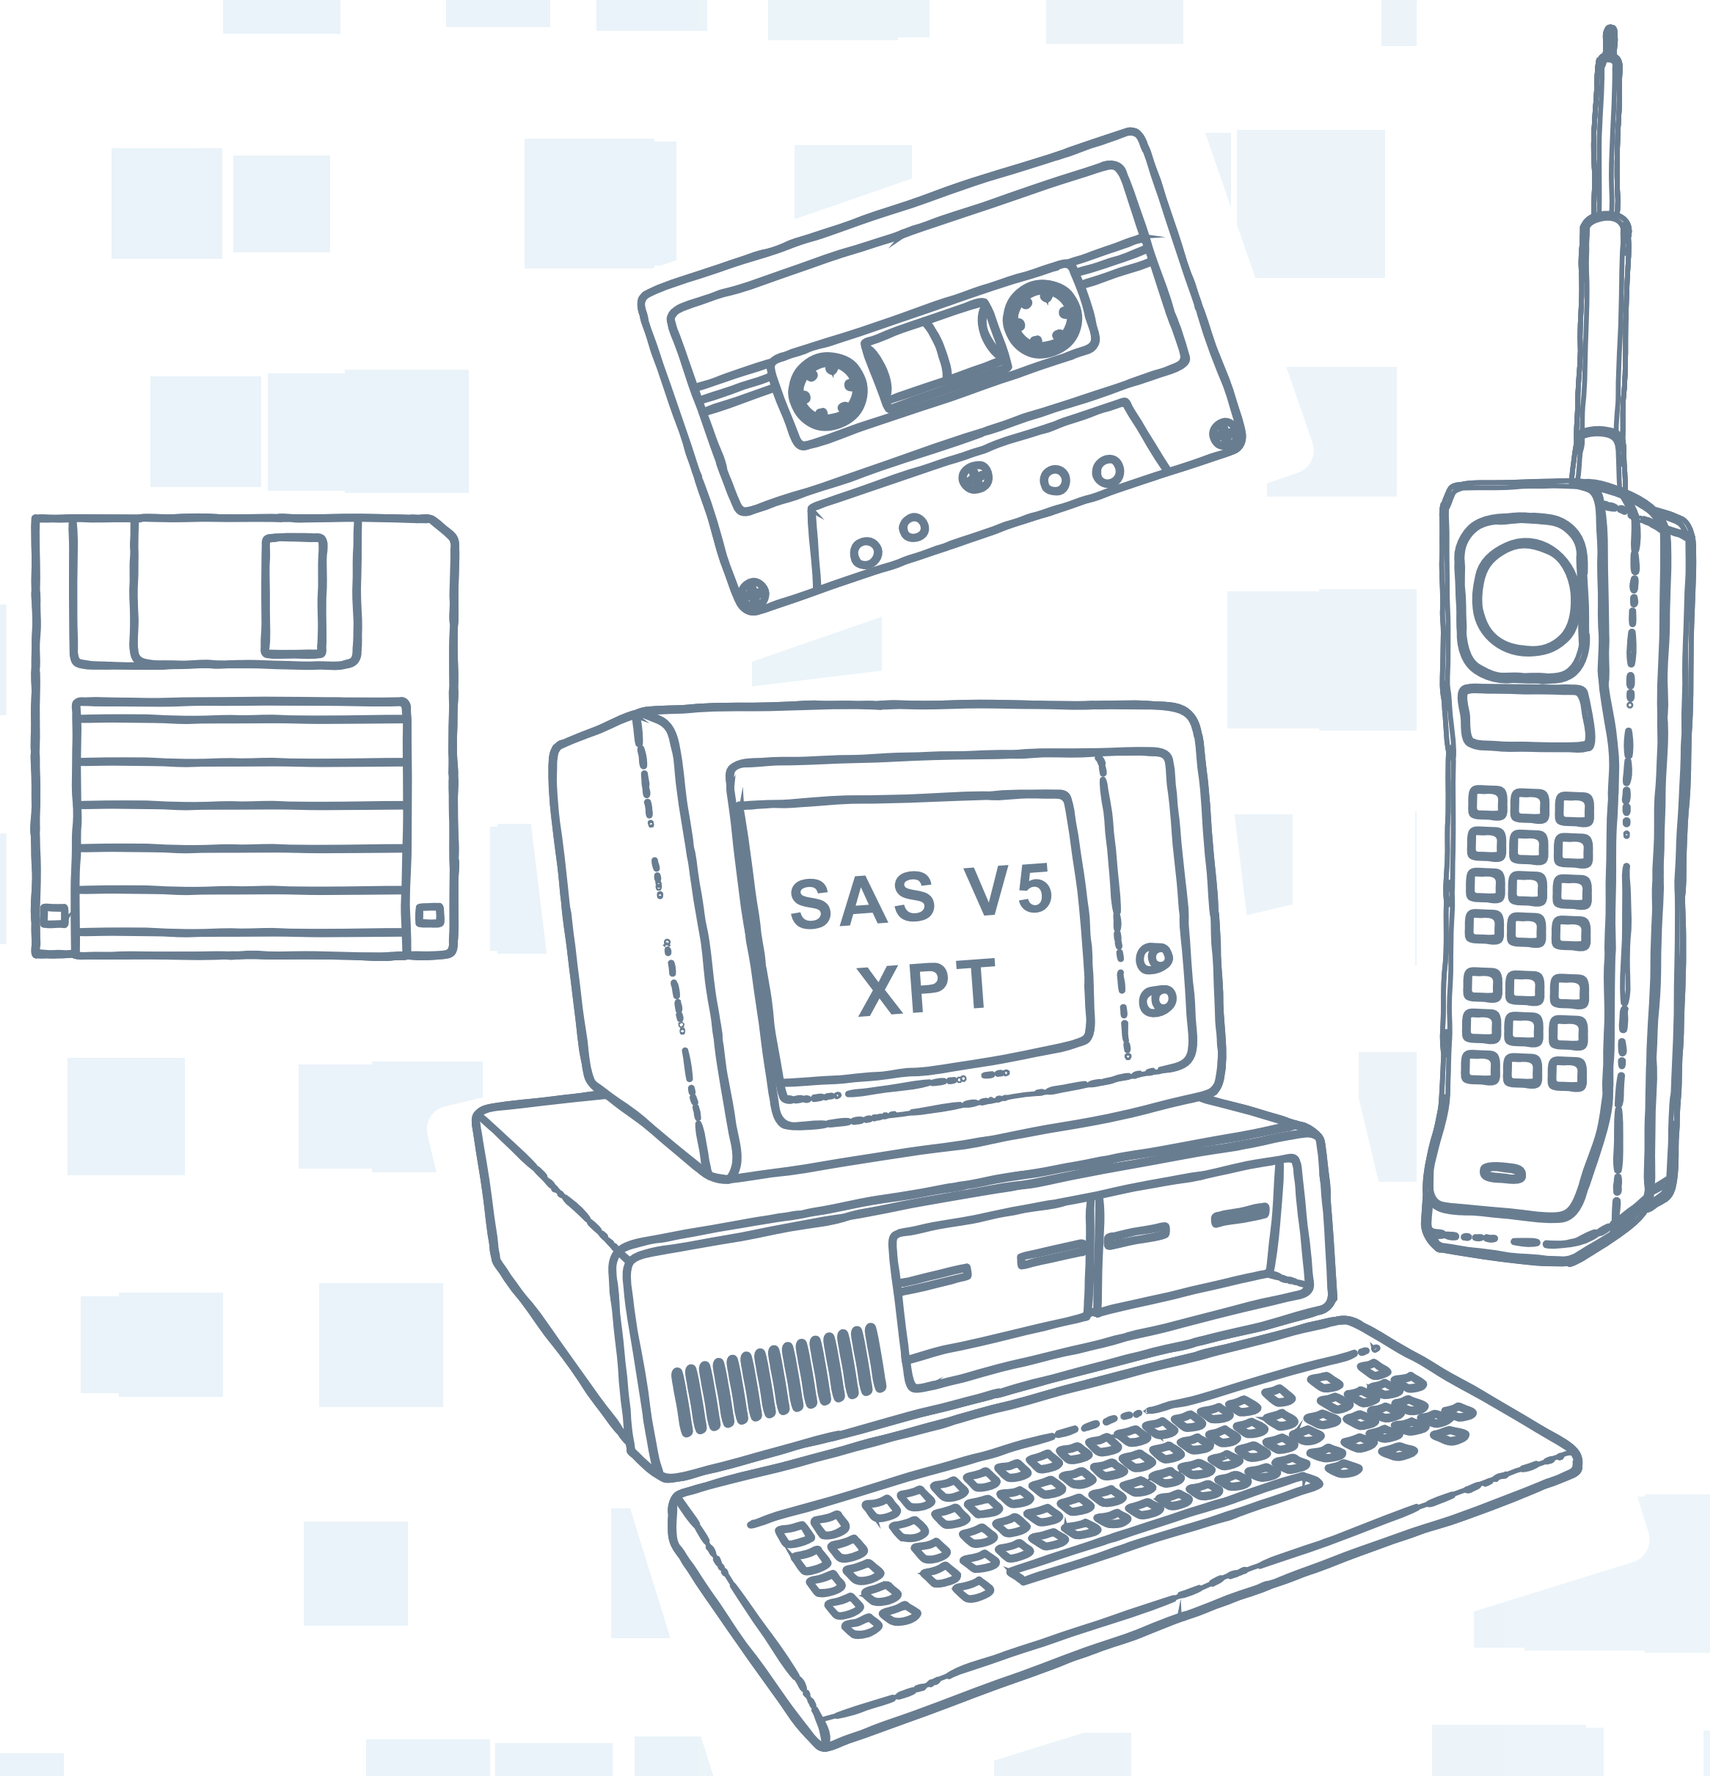

SAS V5
XPT

Submission of a 30-year-old SAS transport format has driven standards definition, tool creation, and processes. How absurd is that?

"Instead of twisting data to fit into rigid, disconnected formats, we must learn to fit analytics directly to the data as captured. This is how analytics are applied throughout the modern world – why not research too?"

Wayne Kubick, CTO HL7

IMAGINE WHAT IT COULD BE…

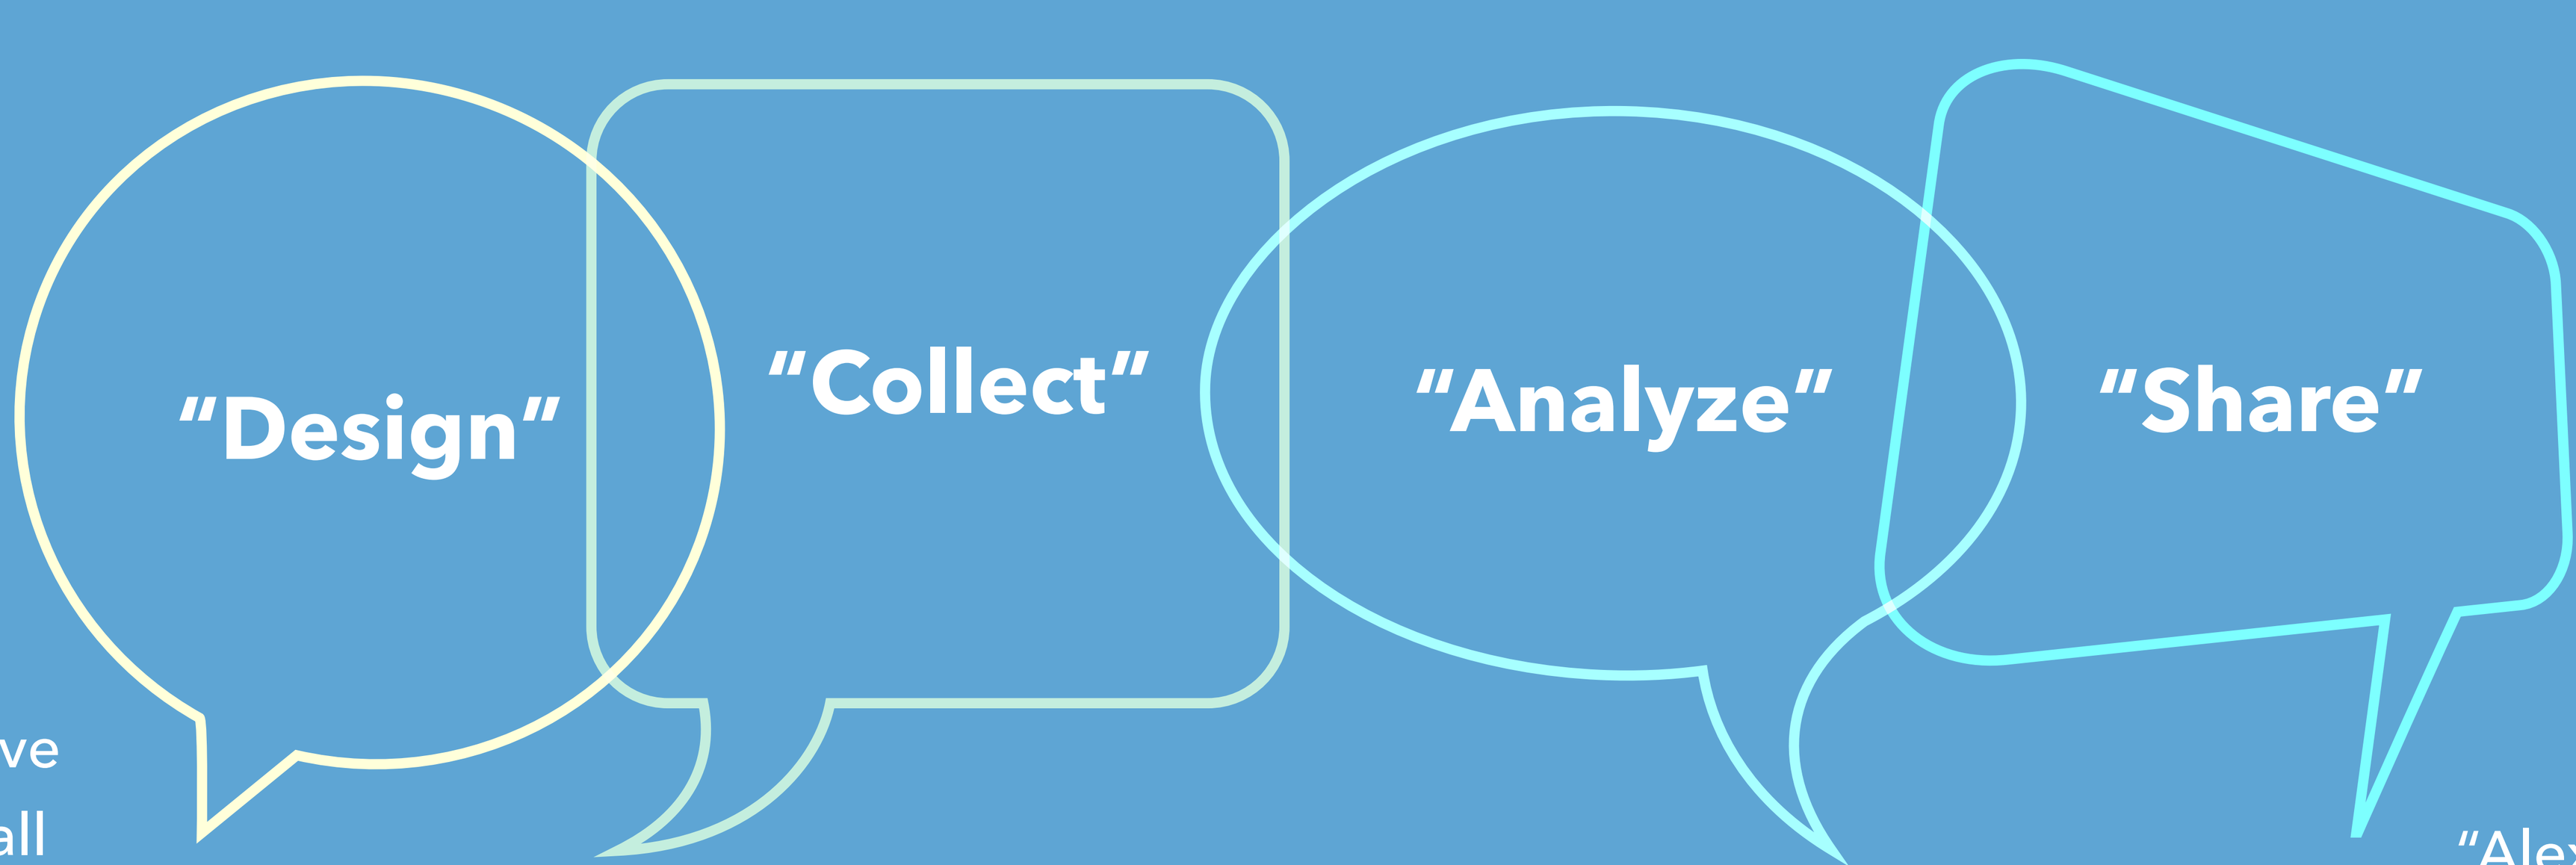

"Design"
"Collect"
"Analyze"
"Share"

Medical Director:
"Alexa, what are the top five exploratory objectives in all active breast cancer trials?"

Clinical Research Associate:
"Alexa, what data is missing for Thalia's week 12 visit?"

Statistician:
"Alexa, is there a statistically significant difference in progressive disease between our drug and the comparator?"

FDA Reviewer:
"Alexa, show me the methods used to confirm my drug reduces the occurrence of progressive disease relative to the comparator."

d-wise

TINYURL.COM/Y92KUJU9