SDTM: Model or Implementation Guide

— Michael Wise, Soumya Rajesh, Kapila Patel

Abstract:

CDISC has developed the SDTM Model to standardize clinical trial data, expediting approval of therapeutic agents. The Implementation Guide (IG) provides navigation for increasingly complex paths from collecting data to mapping those data within a standard framework. Along with the IG, Pinnacle 21 has developed a suite of ubiquitous tools to assess CDISC compliance. Less appreciated, however, is the distinction between Model and IG: the IG is not a mirror of the Model — but rather an interpretation. Pinnacle 21 tools, based on these interpretations, may sometimes suggest unnecessary changes. This poster presents the distinction between Model and IG and provides guidelines to create model compliant databases.

Introducing the Players

- **Model:** Standard model for organizing and formatting data for human and animal studies.
- **Implementation Guide:** Guides the organization, structure and form of that data.
- **Pinnacle 21 Tools:** Based on the Implementation Guide and FDA input, widely used to validate SDTM compliance.

SDTM Interpretation Guidelines

- If the Implementation Guide departs from the Model, the end result will be a dataset that was compliant with the SDTM Model being changed to satisfy the SDTM Implementation Guide.
- Remember that the Implementation Guide merely guides industry through the process of being compliant with the Model.
- Some Laboratory values, "Absolute Neutrophil Count" and "Urine Specific Gravity" for example, are not associated with units.

SD0026	FDAC154	Missing value for --ORRESU, when --ORRES is provided	Original Units (--ORRESU) should not be NULL, when Result or Finding in Original Units (--ORRES) is provided.

Examples:

1. **Study designs could have Disposition or milestone dates that may be misinterpreted**

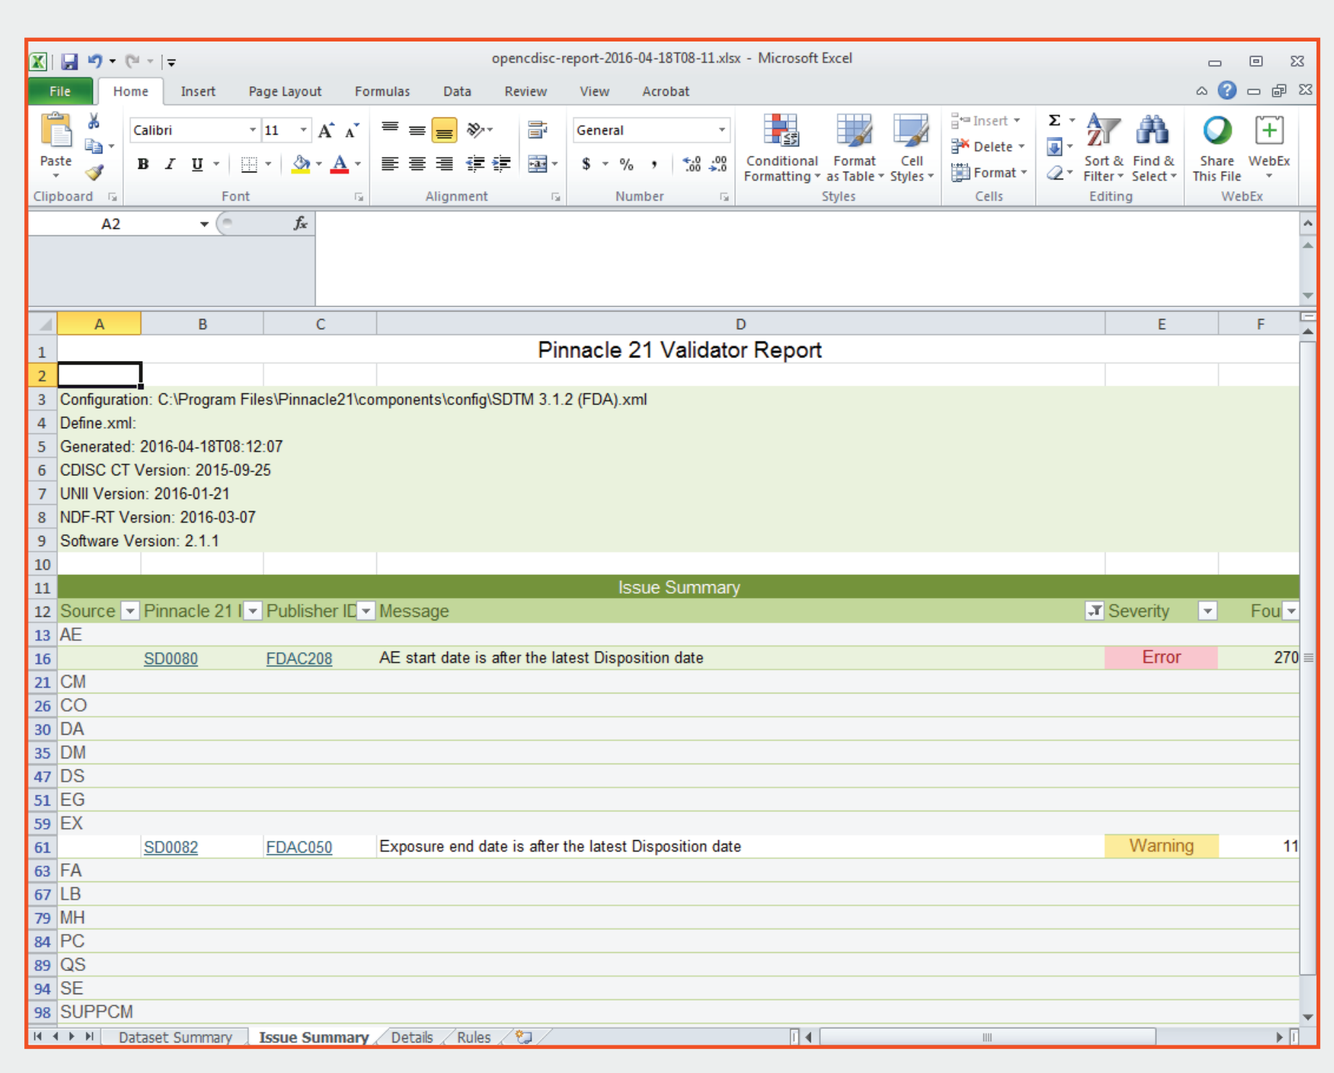
Pinnacle 21 Validator Report
AE start date is after the latest Disposition date
Error
Exposure end date is after the latest Disposition date
Warning

2. **False alarm about discrepancy between AE date & last Disposition date**

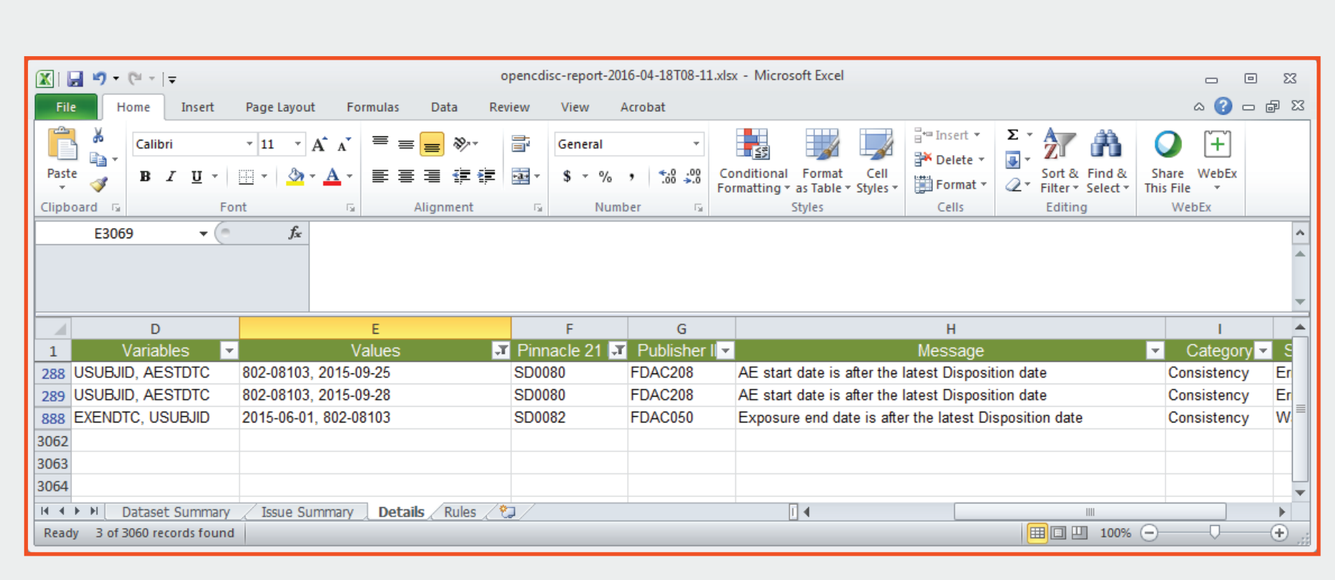
AE start date is after the latest Disposition date
Exposure end date is after the latest Disposition date

3. **AE occurring beyond blinded treatment**

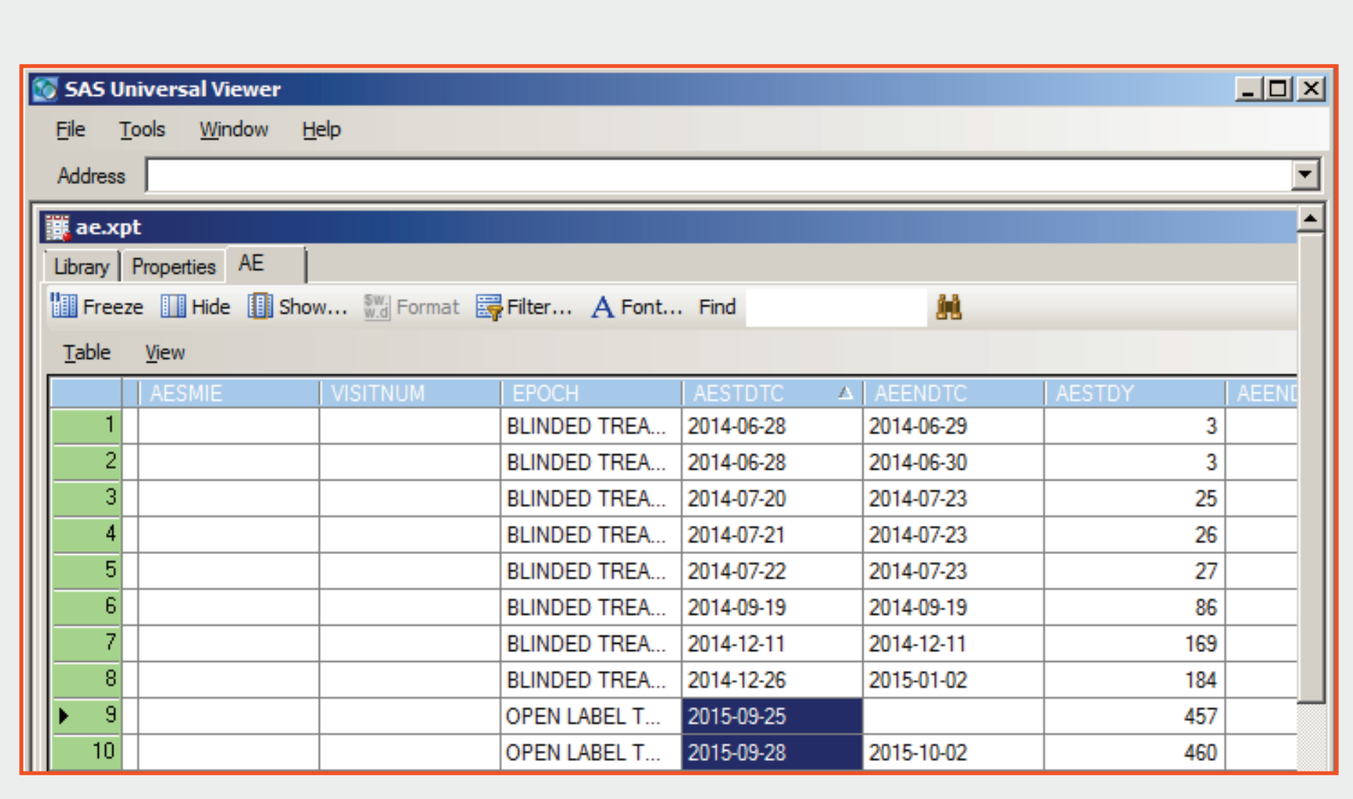
SAS Universal Viewer
ae.xpt

4. **Open-label follows last dose date in blinded treatment**

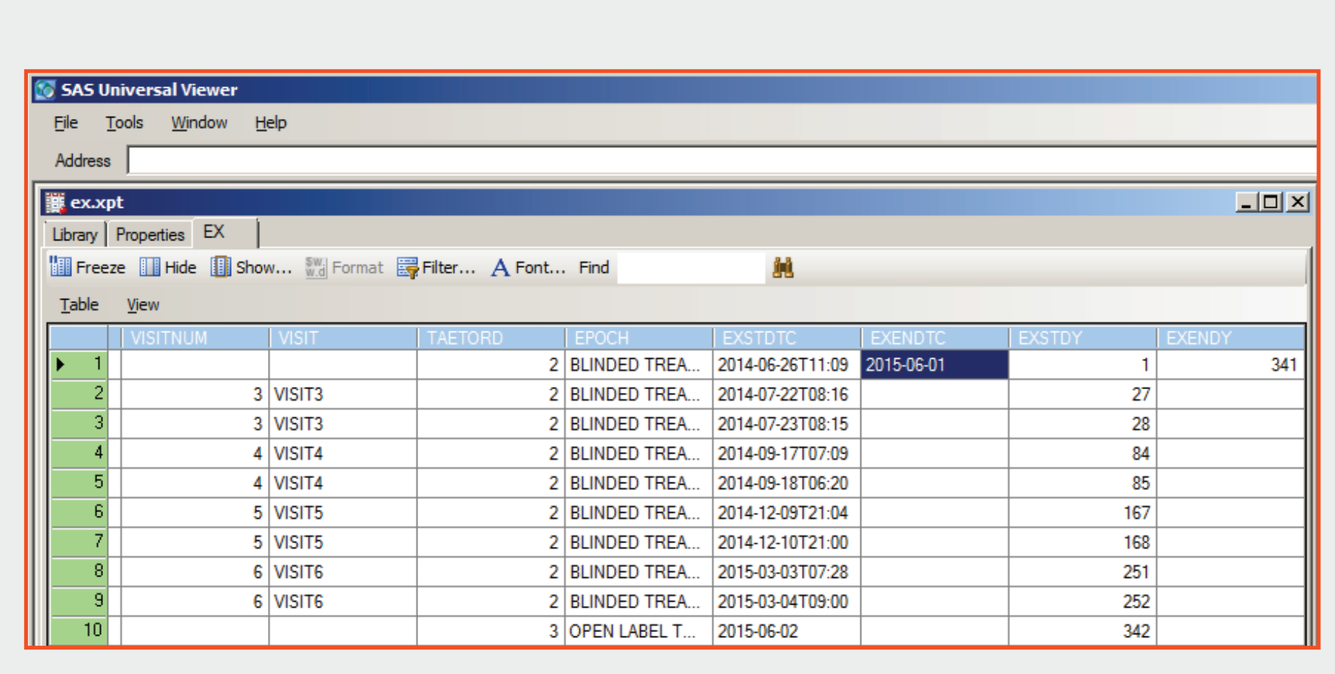
SAS Universal Viewer
ex.xpt

5. **Last date in Disposition**

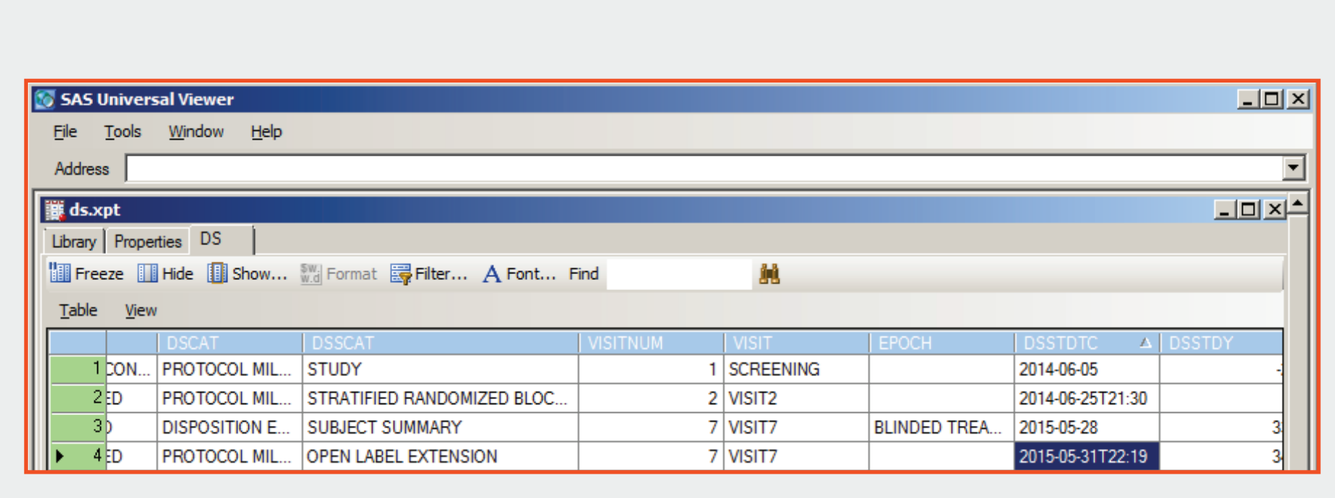
SAS Universal Viewer
ds.xpt

Pinnacle 21 Validation Scores

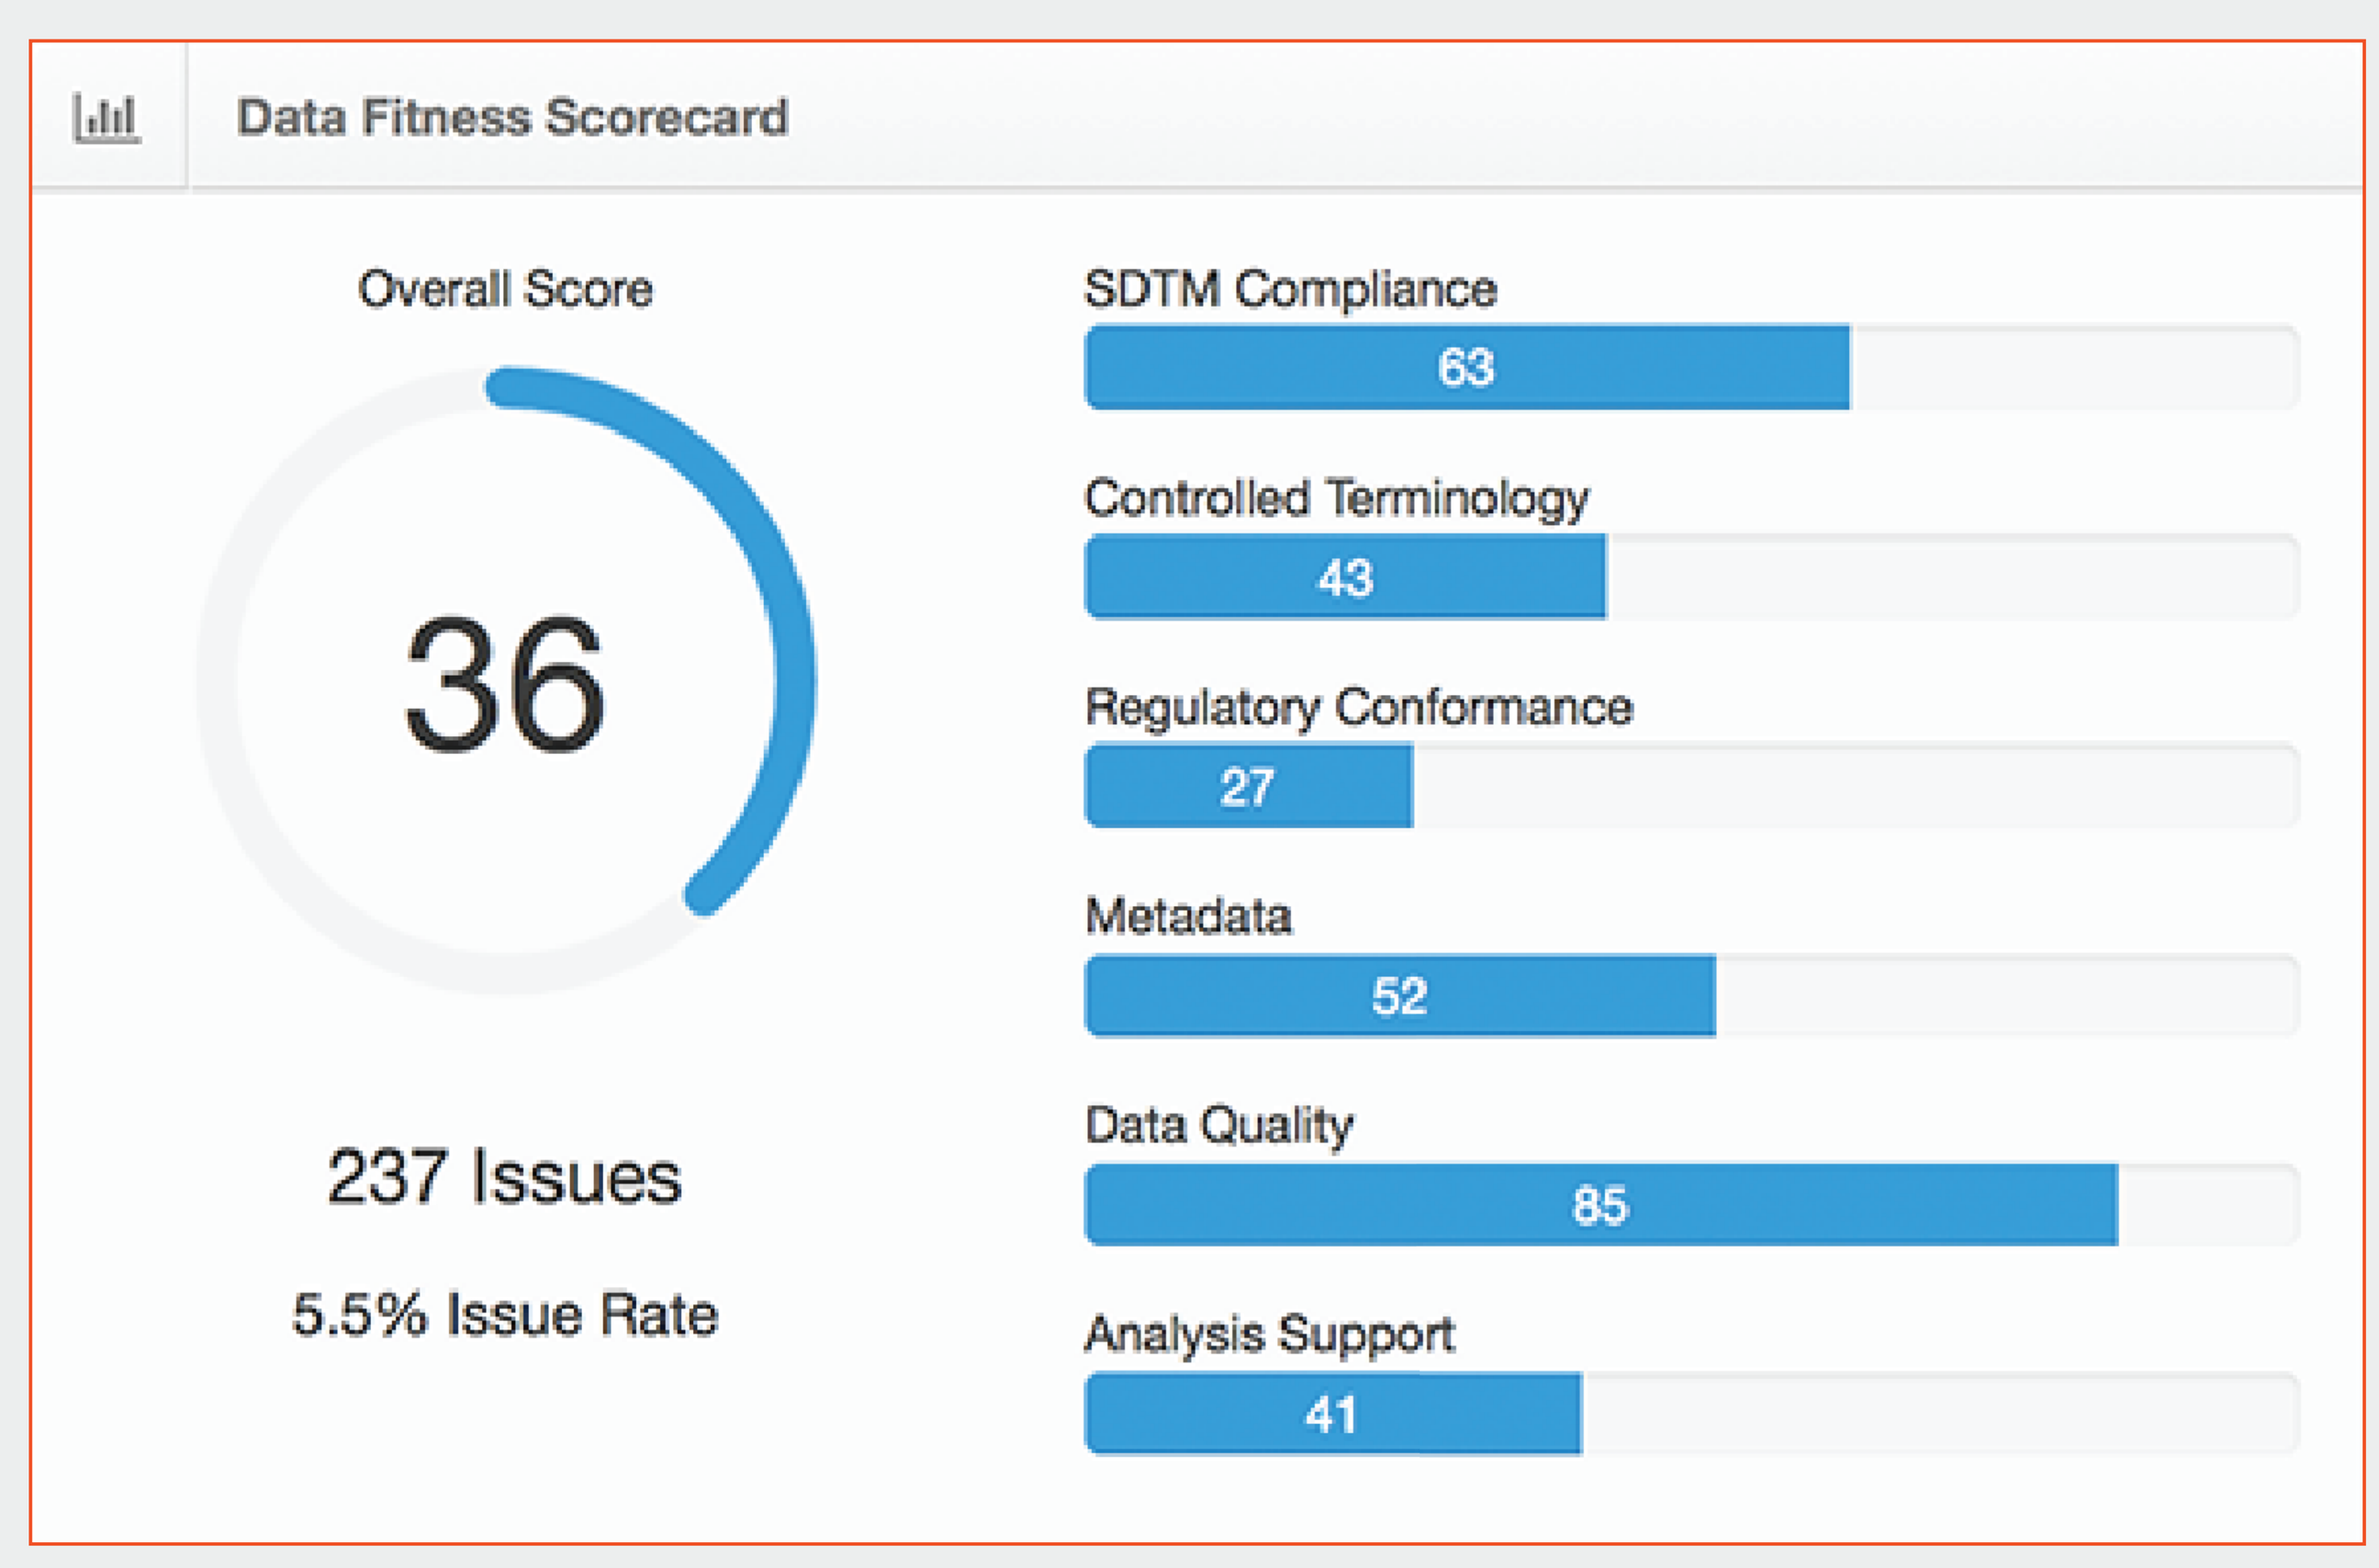
Data Fitness Scorecard
Overall Score
36
237 Issues
5.5% Issue Rate
SDTM Compliance
63
Controlled Terminology
43
Regulatory Conformance
27
Metadata
52
Data Quality
85
Analysis Support
41

Who to Follow:

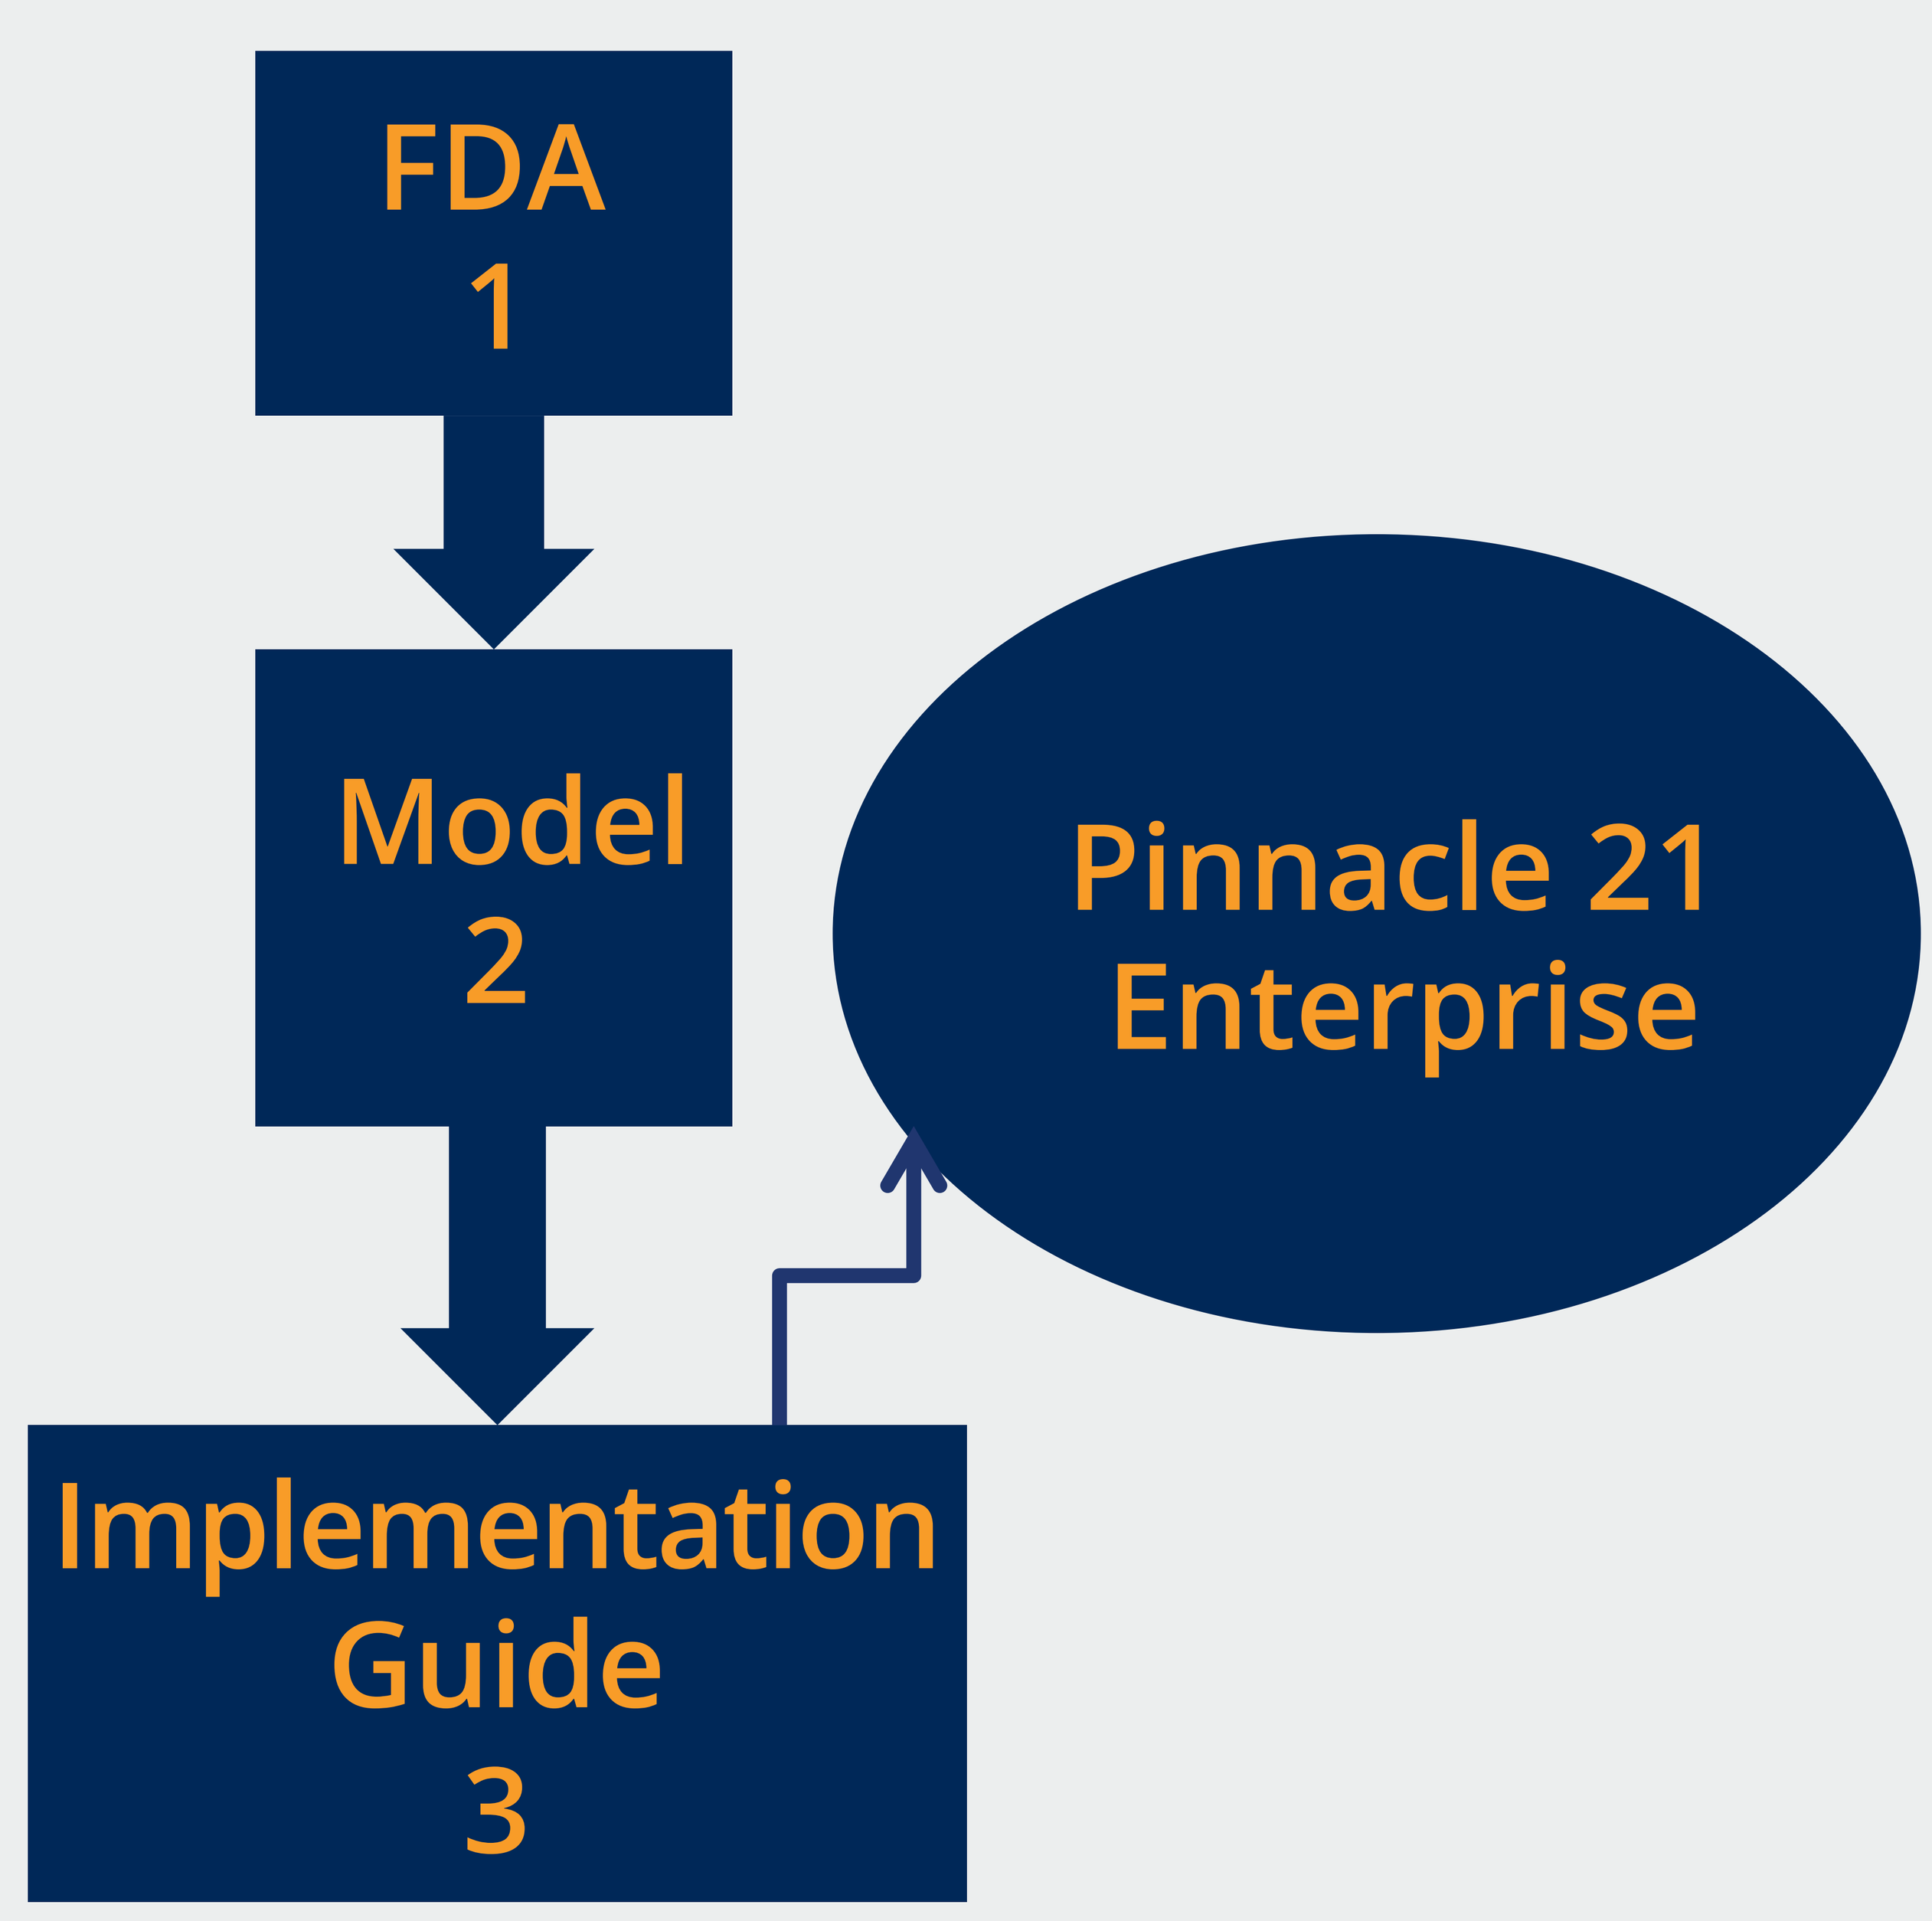
FDA
1
Model
2
Implementation Guide
3
Pinnacle 21 Enterprise

FDA Preferences

- Although not required by the model, the Technical Conformance Guide from FDA states that "EPOCH should be included for clinical subject-level observation"
- The Model permits or expects certain variables be null if others variables are collected. FDA reviewers, however, may request a sponsor to have values for both variables for a given study to be reviewed.

Summary:

When creating an SDTM compliant database it is important to be aware of the rules of the Model and realize that the Implementation Guide is an interpretation of these rules, that could be variable depending on the study design and data collected. It is important to remember the hierarchy of whom to follow to facilitate review and expedite the approval process of a submission.

© 2018 Syneos Health™ All rights reserved

Syneos™ Health